Development and Optimization of LNP Formulation with a Novel Liver-Targeted Ionizable Lipid FL-2266T

Keiko Makita-Suzuki, Noriyuki Kasagi, Shintaro Tanabe, Masashi Yoshimura, Naoto Nakamura, Yuichi Funase, Taisuke Endo, Masahiko Yamamoto, Hirofumi Fukunaga, Masaki Noro, Shigetomo Tsujihata

FUJIFILM Corporation, Kanagawa, Japan

Poster ID 25

FUJIFILM

Abstract

Lipid nanoparticles (LNPs), which have already been used in a commercial siRNA therapeutic, can be regarded as a leading option for mRNA therapeutics vehicles. The key component of LNPs is the ionizable lipid and there are several reports which identified efficient and tolerable ionizable lipids for mRNA therapeutics. However, the lack of availability of ionizable lipids and expertise in LNP GMP production still limits the therapeutic application of mRNA therapies.

To address this, with a rational medicinal chemistry approach, we have synthesized more than three hundred ionizable lipids and evaluated using bioluminescence with firefly luciferase (FLuc) mRNA. LNPs loaded with mRNA encoding FLuc, were administered to ICR mice via intravenous injection. Among these lipids, a novel lipid FL-2266T, which contains di-amino head with biodegradable ester-containing branched tail structure showed preferable luciferase expression. Particle size and encapsulation efficiency (EE) of FL-2266T LNPs were 85.2 nm and >98%, respectively (Table 1). FL-2266T showed almost 2-fold higher luminescence in the liver than DLin-MC3-DMA (MC3) as a benchmark (Figure 1). Furthermore, FL-2266T displayed the rapid elimination from plasma and tissue, in contrast to MC3 (Figure 2).

Next, we formulated FL-2266T LNPs loaded with human erythropoietin (hEPO) mRNA and evaluated their performance in ICR mice. Our LNPs showed 2-fold higher hEPO production compared with MC3 LNPs (Figure 3).

FL-2266T LNPs induced the acute and transient expression of proinflammatory cytokines, which is known as a common effect among nanoparticles. The levels in plasma induced by FL-2266T LNPs were comparable to those of MC3 [ref1] (Figure 4).

Now, we have installed NanoAssemblr® GMP system to our liposome & LNP-dedicated GMP facility and are ready to offer our CDMO service from lab-scale to commercial manufacturing of LNP formulation based on the partnership with Precision NanoSystems Inc (Figure 5). FL-2266T is available for formulation development in our CDMO service.

Characterization of LNPs

Table 1. Characterization of LNPs

Lipid	Size (nm)	ζ potential (mV) pH 7.4	ζ potential (mV) pH 5.5	EE (%)
MC3	83.6	-7.8	20.1	97.3
FL-2266T	85.2	-8.3	24.2	> 98

Reference

1. Maugeri, M., Nawaz, M., Papadimitriou, A. *et al.* Linkage between endosomal escape of LNP-mRNA and loading into EVs for transport to other cells. *Nat Commun* **10,** 4333 (2019).

Luciferase expression in mice

Figure 1. Whole body imaging by luminescence

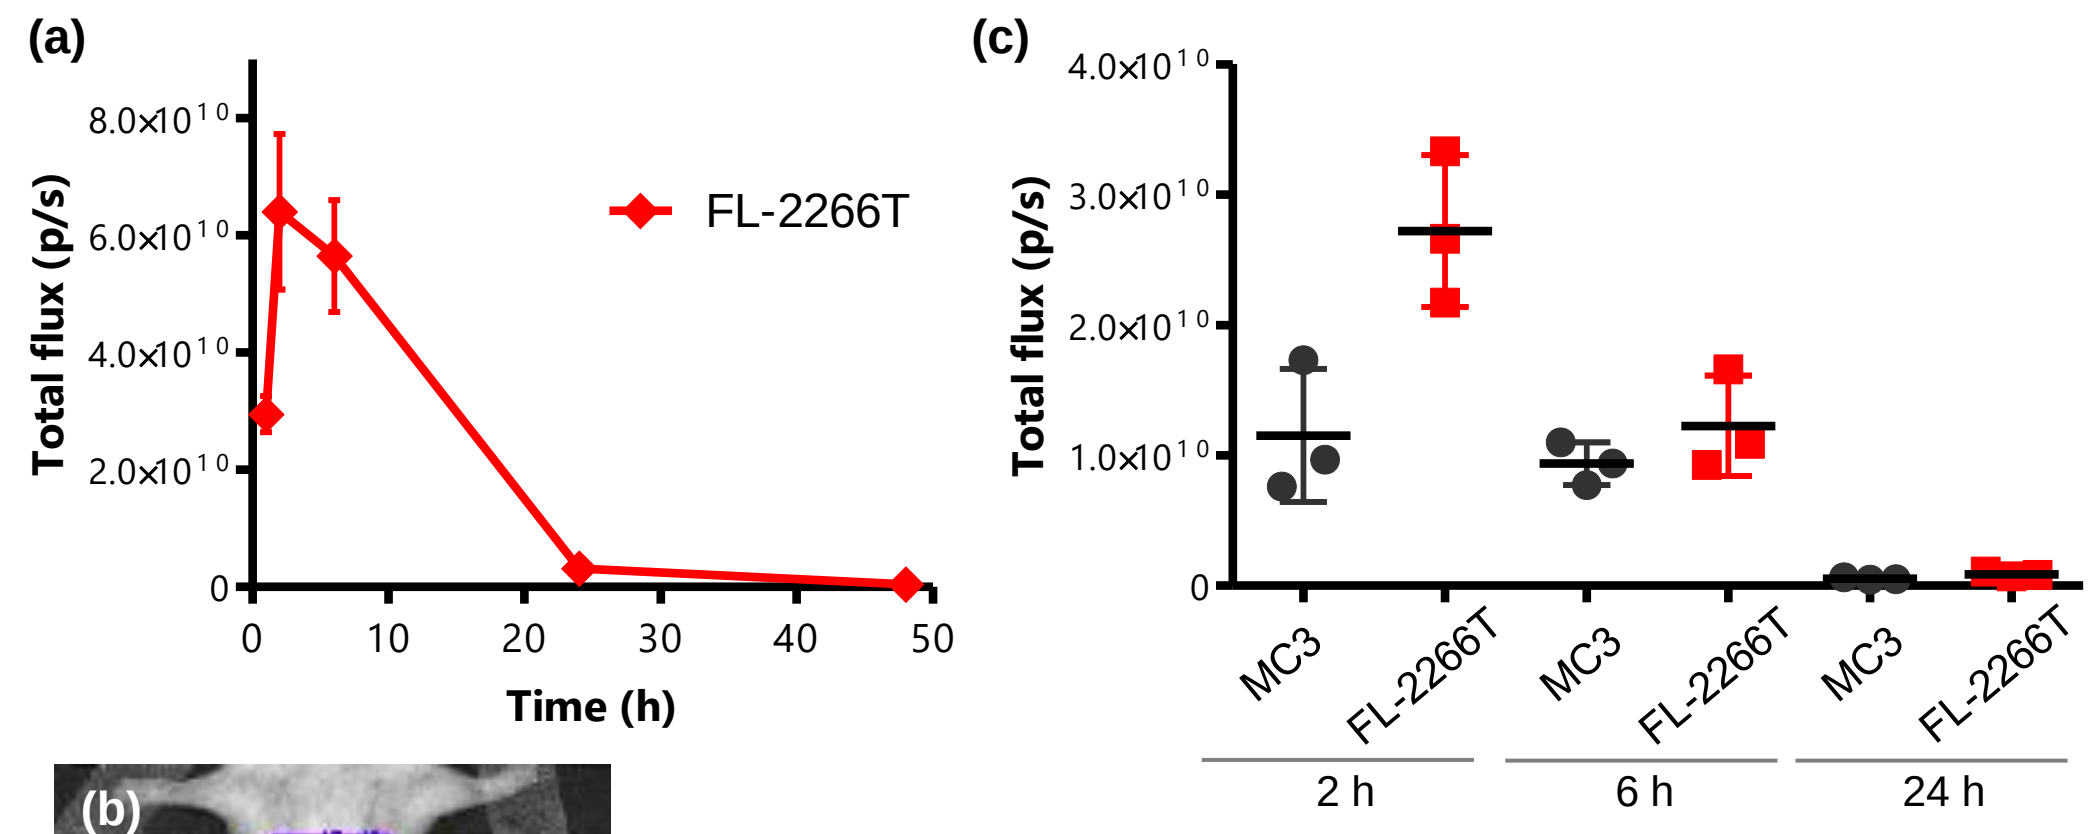
(a)
Total flux (p/s)
FL-2266T
Time (h)
(c)
Total flux (p/s)
MC3
FL-2266T
2 h
6 h
24 h

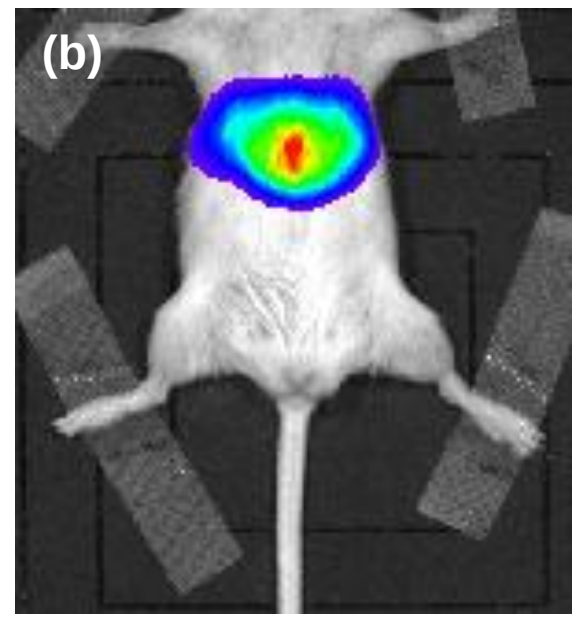
(b)

A single dose of FL-2266T or MC3 LNPs loaded with FLuc mRNA was administered to ICR mice at 0.2 mg/kg dose FLuc mRNA.
(a) Time course of whole body luminescence of FL2266T (N = 3, mean +/- SD)
(b) Typical image of whole body luminescence of FL-2266T at 6 h post injection
(c) Ex vivo luminescence of the liver at 2, 6, and 24 h post injection (N = 3, mean +/- SD)

- FL-2266T was identified through in vivo luminescence assay.
- FL-2266T LNPs showed almost 2-fold higher luminescence in the liver than MC3.

FL-2266T LNPs biodistribution in mice

Figure 2. Lipid pharmacokinetic profiles in murine plasma and tissue

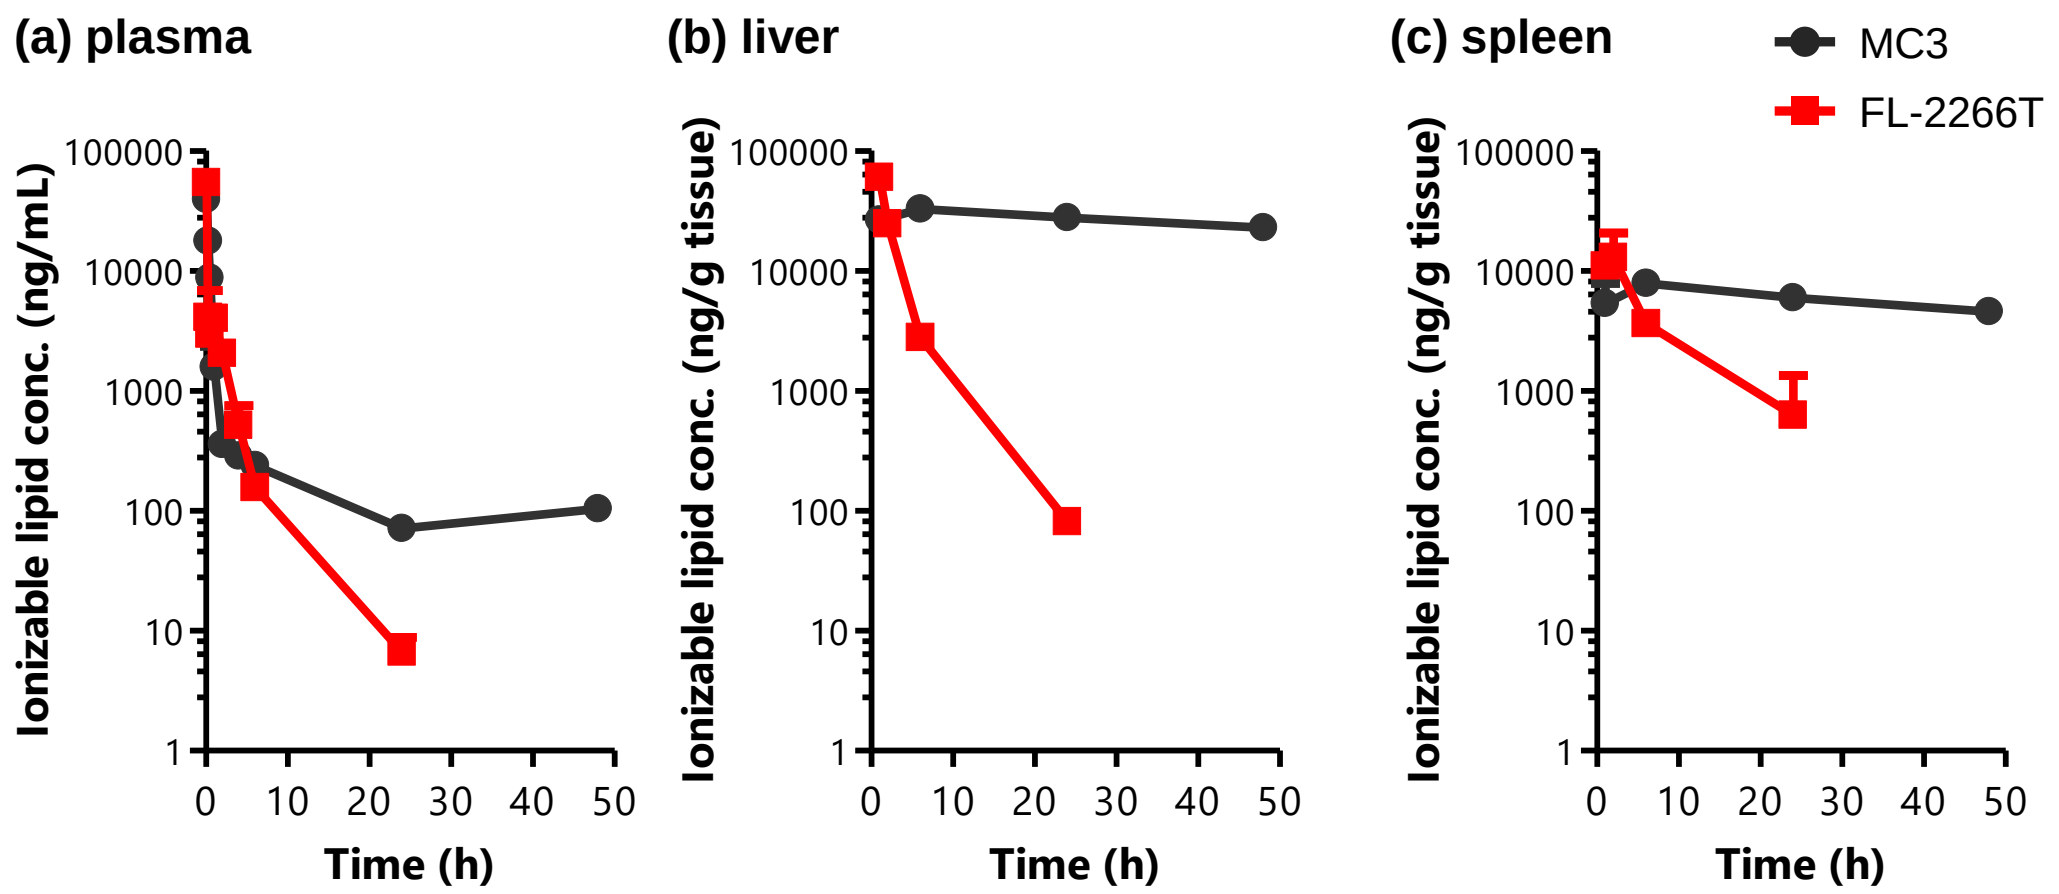
(a) plasma
(b) liver
(c) spleen
MC3
FL-2266T
Ionizable lipid conc. (ng/mL)
Ionizable lipid conc. (ng/g tissue)
Time (h)

A single dose of FL-2266T or MC3 LNP loaded with FLuc mRNA was intravenously administered to ICR mice at 0.2 mg/kg FLuc mRNA After single injection, concentration–time profiles of ionizable lipids in plasma (a), liver (b), and spleen (c) were determined using LC-MS/MS. (N = 3 per time point, mean + SD)

- FL-2266T displayed the rapid elimination from plasma and tissue compared to MC3.
- The liver tissue half-life of FL-2266T was calculated as 2.88 hours, in contrast to 81.0 hours half-life of MC-3.

hEPO production in mice

Figure 3. hEPO protein levels in murine plasma at 6 h

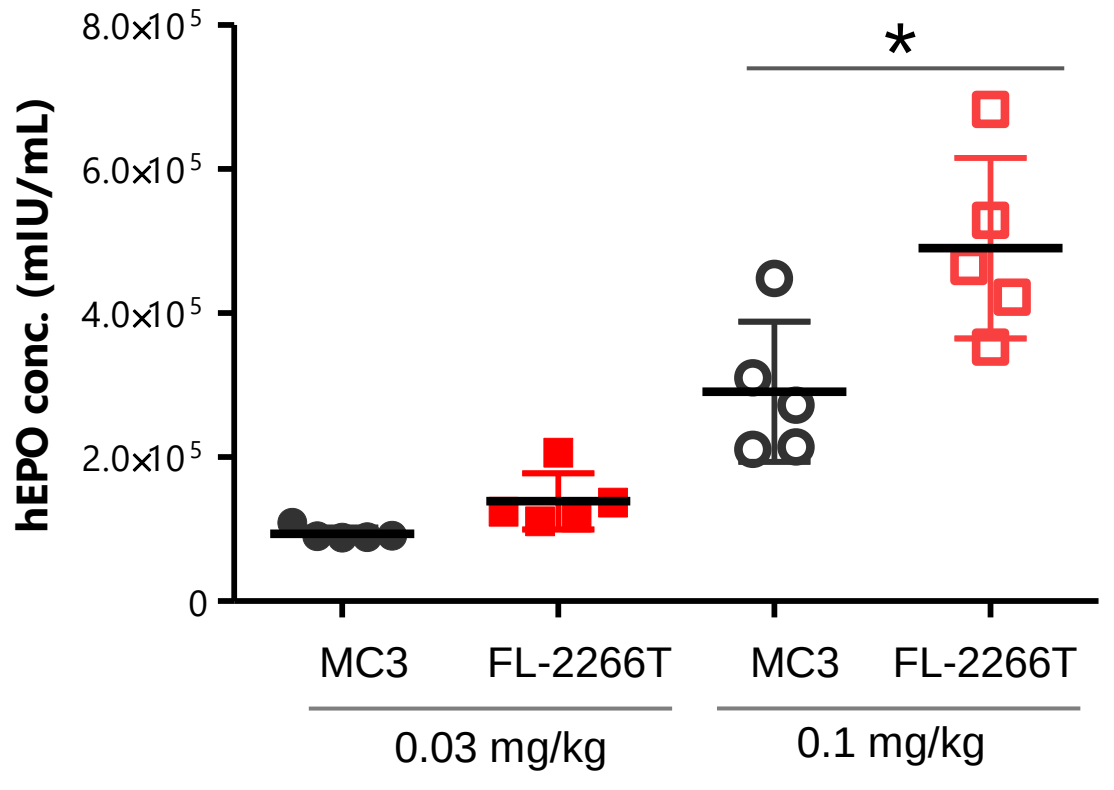
hEPO conc. (mIU/mL)
*
MC3
FL-2266T
MC3
FL-2266T
0.03 mg/kg
0.1 mg/kg

A single dose of FL-2266T LNP or MC3 LNP loaded with hEPO mRNA was intravenously administered to ICR mice at 0.03 and 0.1 mg/kg as hEPO mRNA. At 6 h post injection, hEPO protein levels in plasma were determined by ELISA. (N = 5 per group, mean +/- SD, * P<0.05)

- FL-2266T LNPs showed higher hEPO expression than MC3 LNPs at both 0.03 and 0.1 mg/kg.
- The hEPO levels at 0.1 mg/kg of FL-2266T LNPs were almost 2-fold higher than those of MC3.
- Dose-dependent hEPO production was observed from 0.03 to 0.1 mg/kg.

Inflammatory cytokines in mice

Figure 4. Pronflammatory cytokines in mice

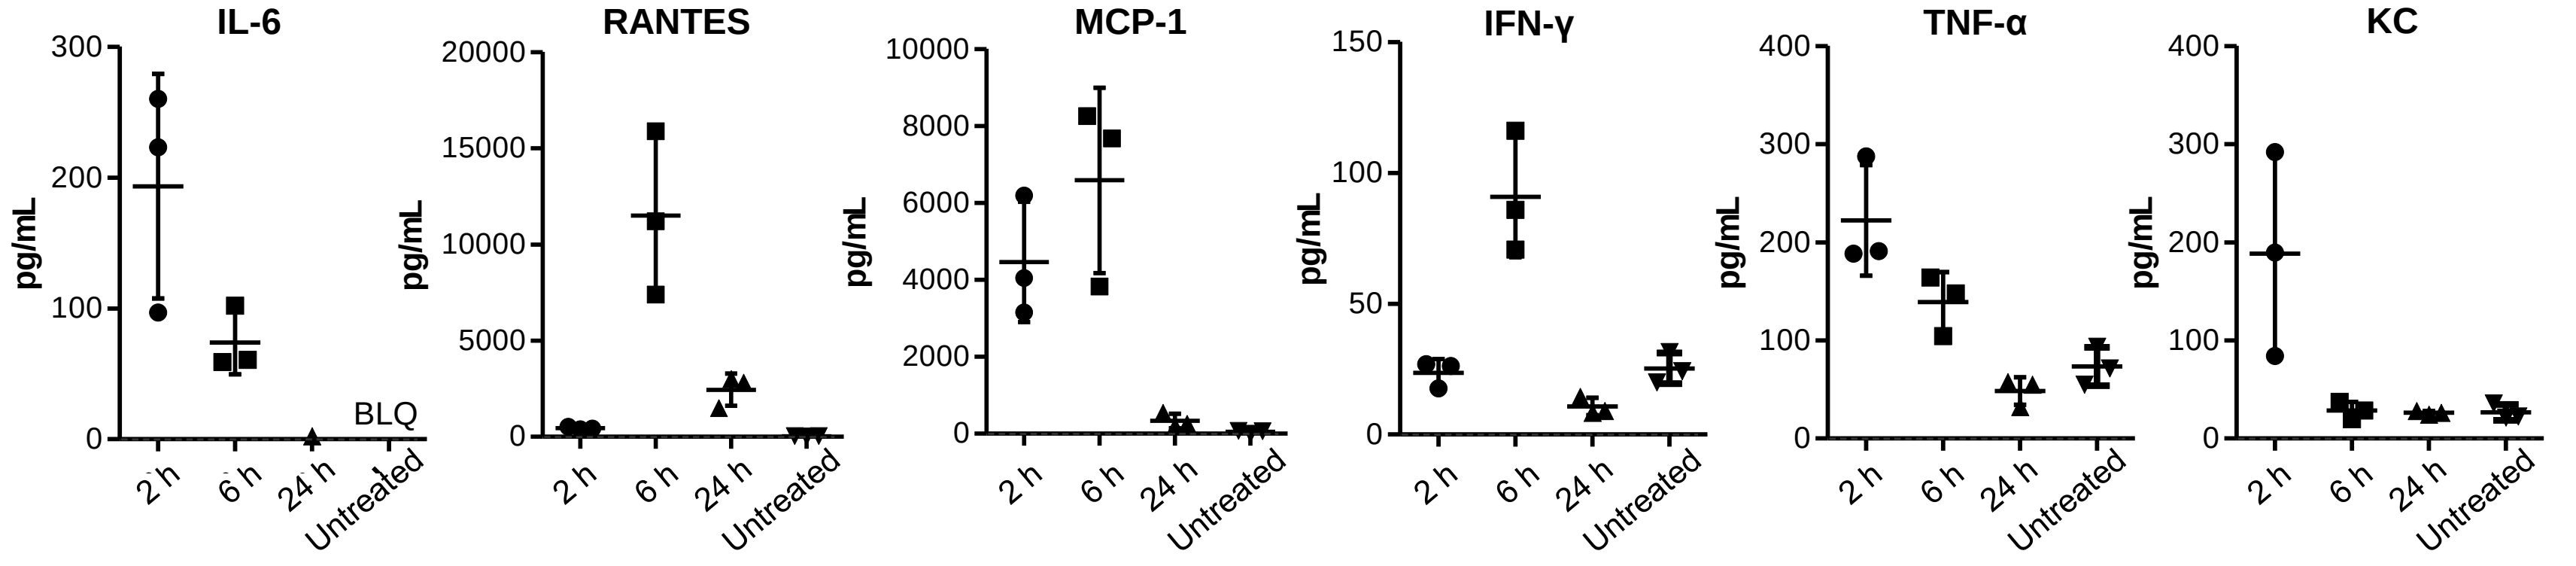
IL-6
RANTES
MCP-1
IFN-γ
TNF-α
KC
pg/mL
BLQ
2 h
6 h
24 h
Untreated

A single dose of FL-2266T LNP loaded with FLuc mRNA was intravenously administered to ICR mice at 0.2 mg/kg FLuc mRNA. Cytokines levels in plasma at 2, 6, and 24 h post injection were determined using Bio-plex system. (N = 3 per group, mean +/- SD, BLQ data points were not shown in graphs. BLQ: All data points were BLQ)

- FL-2266T LNPs caused expression of proinflammatory cytokines, such as IL-6, RANTES, MCP-1, IFN-γ, TNF-α, and KC.
- The expression of cytokines was acute and transient.
- The cytokine levels in plasma induced by FL-2266T LNPs were comparable to those of MC3 previously reported [ref1].

CDMO capability

Figure 5. FUJIFILM CDMO service partnered with Precision NanoSystem Inc.

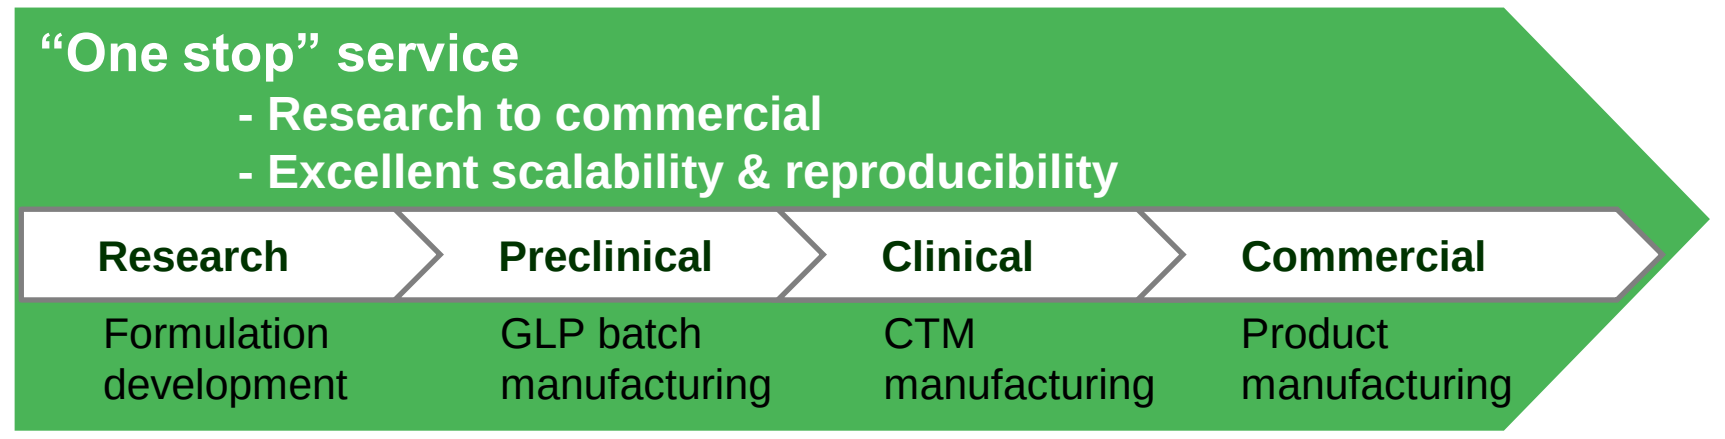
"One stop" service
- Research to commercial
- Excellent scalability & reproducibility
Research
Formulation development
Preclinical
GLP batch manufacturing
Clinical
CTM manufacturing
Commercial
Product manufacturing

	Ignite™	Blaze™	GMP system
NanoAssemblr® platform	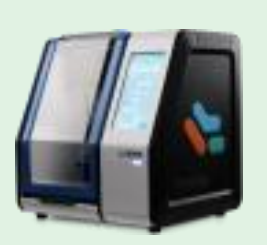	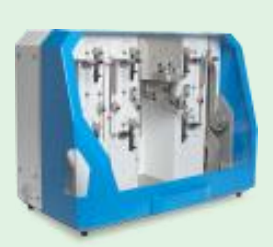	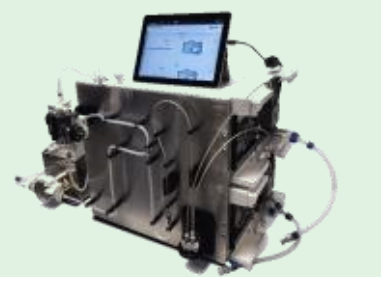
Development phase	Research to preclinical	Preclinical	Preclinical to commercial
Scale	1 to 15 mL	10 to 1000 mL	0.2 to >100 L
Facility	Non-GMP	Non-GMP	GMP

- GMP system will be ready in new facility dedicated for LNP and Liposome at the end of 2020.
- FUJIFILM original lipids including FL-2266T can be available for formulation development.

Summary

- FUJIFILM ionizable lipid FL-2266T shows high protein expression via intravenous injection.
- FL-2266T displays the rapid elimination from plasma and tissue.
- FL-2266T is available for formulation development in CDMO service.

8th International mRNA Health Conference/Nov 9-10, 2020