Design and Devepolment of Novel Ionizable Lipid Nanoparticle, and its Evaluation as pDNA Delivery Vector

〇Tatsuya Nitabaru, Naoto Nakamura, Yusuke Totsuka, Masafumi Nishino, Masahiko Yamamoto, Shintaro Tanabe, Koki Endo, Shigetomo Tsujihata, Hirofumi Fukunaga

FUJIFILM corporation

FUJIFILM

11th international mRNA health conference
2023/10/31-11/02, Berlin

Abstract

Lipid nanoparticles (LNPs) have attracted a lot of interest as one of the most validated pathways to realize nucleic acid therapeutics, highlighted by the successful commercialization of siRNA therapeutics, Onpattro™, and mRNA vaccines, Spikevax™ and Comirnaty™. However, the translation of LNPs from preclinical research to practical therapeutic use still poses numerous challenges such as large scale synthesis, stability and purity of ionizable lipids, formulation optimization, in vivo efficacy-safety balance, and potency translation across species. We have been developing our proprietary ionizable lipid library and LNP formulations since before the COVID-19 pandemic. Two of our first-generation ionizable lipids are available as GMP-grade products and one is currently used in a self-amplifying RNA (saRNA) vaccine clinical trials in Japan. Currently, our efforts are focused on synthesizing a second-generation library aimed at expanding the scope of applicable nucleic acid modalities and therapeutic applications.

Here, we describe some of our design rules of ionizable lipid structure elucidated through comprehensive in vivo screening and rigorous analysis. First, we have identified a direct correlation between specific physicochemical properties, apparent pKa value of LNP, cLogP value of ionizable lipids, and their impact on in vivo outcomes. Additionally, our analysis has revealed that the each ionizable lipids exerts an influence on formulations. Moreover, our LNP indicates that there is an optimal ionizable lipid for each type nucleic acid. We also describe an application of our ionizable lipids to in vivo pDNA delivery. Our LNP showed higher IgG induction than that of commercially available reagent when locally administered as a vaccine. The best ionizable lipid in pDNA vaccination is different from that in intravenous mRNA delivery, and this mechanism is under analysis.

Design Concept of Novel Ionizable Lipid

Design concept

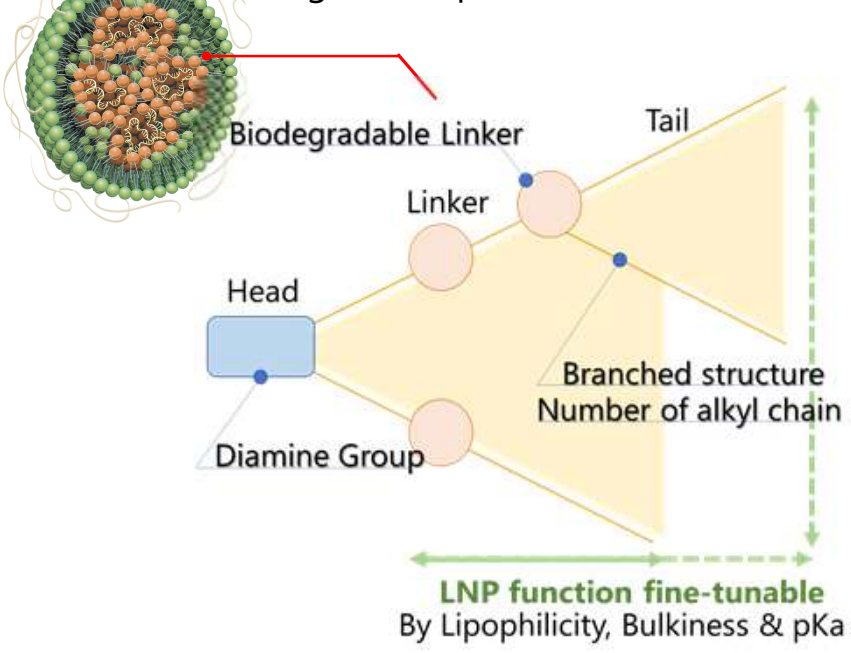
Biodegradable Linker
Tail
Linker
Head
Branched structure
Number of alkyl chain
Diamine Group
LNP function fine-tunable
By Lipophilicity, Bulkiness & pKa

Our design concept features a "head – biodegradable linker – hydrophobic tail" structure, where we employed a diamine moiety as a head part.

Apparent pKa values of LNPs, one of the important parameters for efficient endosomal escape, were precisely controlled by changing both alkyl substituents in the head and lipophilicity or bulkiness in the tail.

Shape of lipid, which is reported to contribute to membrane fusion process, can be controlled by bulkiness of the tail.

Through our structure optimization process, we synthesized more than 400 ionizable lipids and evaluated them in vivo using siRNA (i.v.) and mRNA (i.v./i.m.). We identified several ionizable lipids for further characterization and development.

Summary of Our Ionizable Lipid Library

Name	Type A	Type B	Type C	Type D
Preferred Application	**siRNA** **mRNA** pDNA	**mRNA**	**mRNA**	**pDNA**
Mol. weight Min-MAX	700-1000	700-1500	700-1000	400-1100
clogP Min-MAX	14-25	12-35	15-21	4-25
Apparent pKa of LNPs	4.7-7.2 (partially depends on clogP value of the ionizable lipid)			

- We identified different types of ionizable lipid for each type nucleic acid through in vitro and in vivo screening.

FUJIFILM's Ionizable Lipid & mRNA-LNP Library (Type A-C)

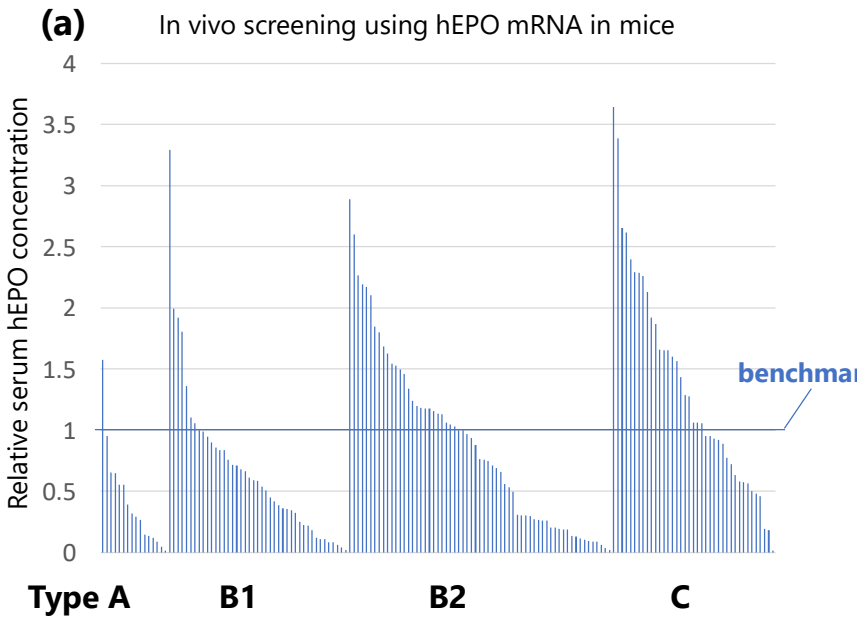
(a)
In vivo screening using hEPO mRNA in mice
Relative serum hEPO concentration
4
3.5
3
2.5
2
1.5
1
0.5
0
benchmark
Type A
B1
B2
C

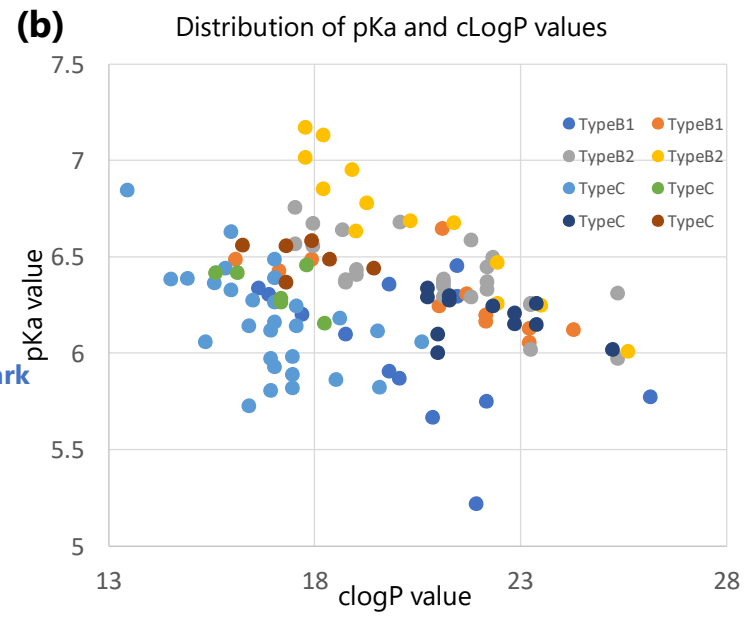
(b)
Distribution of pKa and cLogP values
pKa value
7.5
7
6.5
6
5.5
5
13
18
23
28
clogP value
TypeB1
TypeB1
TypeB2
TypeB2
TypeC
TypeC
TypeC
TypeC

- Our ILs showed higher protein expression than benchmark in mice especially Type B-C.
- Apparent pKa values are controllable by changing clogP (head-tail lipophilicity).

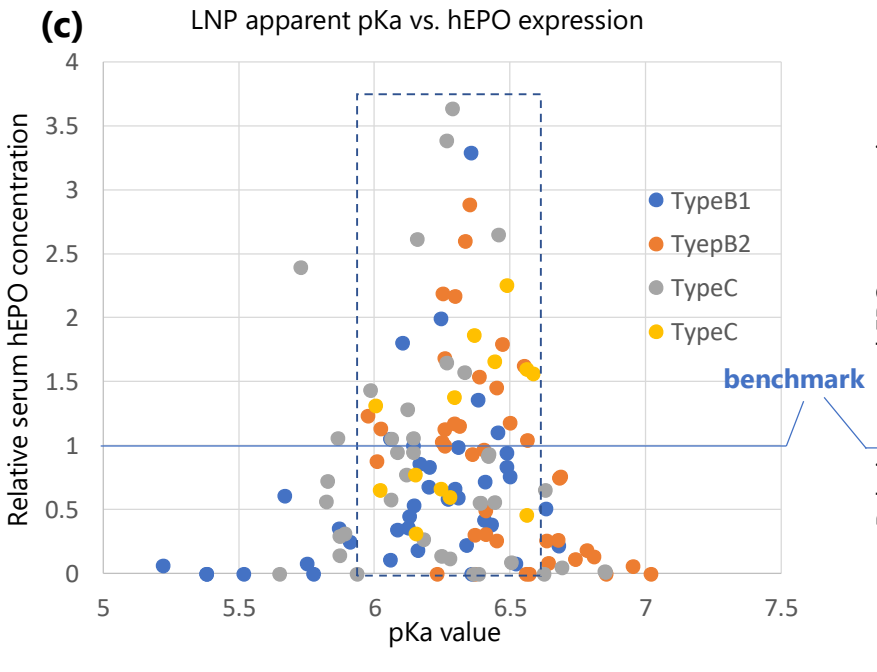
(c)
LNP apparent pKa vs. hEPO expression
Relative serum hEPO concentration
4
3.5
3
2.5
2
1.5
1
0.5
0
5
5.5
6
6.5
7
7.5
pKa value
TypeB1
TyepB2
TypeC
TypeC
benchmark

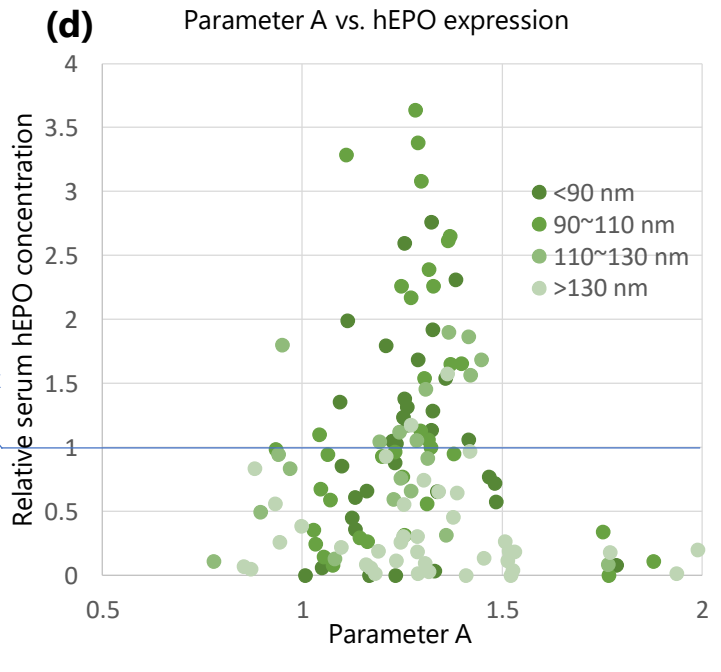
(d)
Parameter A vs. hEPO expression
Relative serum hEPO concentration
4
3.5
3
2.5
2
1.5
1
0.5
0
0.5
1
1.5
2
Parameter A
<90 nm
90~110 nm
110~130 nm
>130 nm

- Optimal apparent pKa values posit 6.0-6.5 regardless of ILs Type, when administered i.v.
- Other parameters control i.v.-administered LNP performance.

Figure 1. Analysis of mRNA-LNPs

(a) hEPO protein expression levels of mRNA-LNP in plasma compared with benchmark LNP, 6 hours after i.v. administration (0.1 mg/kg as hEPO mRNA), (b) Scatter plot of apparent pKa and clogP values of different lipid type, (c) Scatter plot of hEPO protein expression levels and apparent pKa, (d) Scatter plot of hEPO protein expression levels and a certain parameter A

- For rat and NHP data, please check Poster 38

pDNA Transfection in vitro & in vivo (Type D)

Case1. In vitro transfection

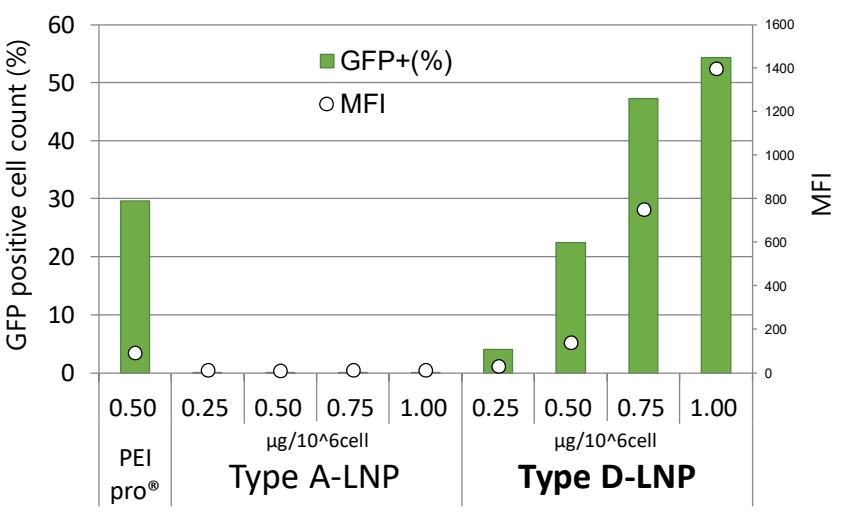
GFP positive cell count (%)
60
50
40
30
20
10
0
GFP+(%)
MFI
1600
1400
1200
1000
800
600
400
200
0
MFI
0.50
0.25
0.50
0.75
1.00
0.25
0.50
0.75
1.00
PEI pro®
µg/10^6cell
Type A-LNP
µg/10^6cell
Type D-LNP

Table. 1 Physicohemical Parameters

Ionizable lipid	diameter (nm)	Zeta potential (mV) pH7.4	Zeta potential (mV) pH5.5	EE* (%)
Type-A	118	-11.2	13.9	99.5
Type-D	88	-7.27	15.9	99.1

*EE; Encapsulation Efficiency

- Type D-LNP showed efficacious pDNA delivery and dose-dependent GFP expression in vitro (HEK293 cells).

Figure 2. Comparison of different LNPs as pDNA vector

GFP expressed cell count (left) and its median fluorescence intensity (right), 3 days after transfection into HEK293T cells of both pDNA-LNPs and pDNA-PEI pro® complex.

Case2. In vivo immunization

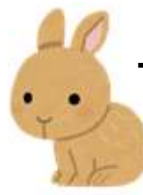

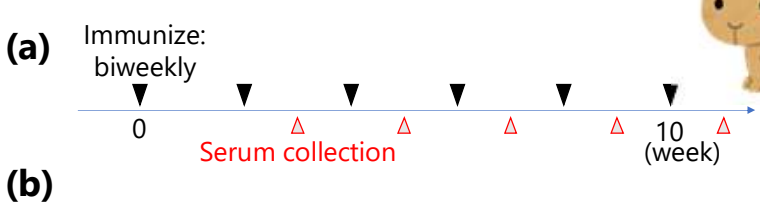
(a)
Immunize:
biweekly
0
Serum collection
10
(week)

Table. 2 Physicohemical Parameters

Ionizable lipid	diameter (nm)	Zeta potential (mV) pH7.4	Zeta potential (mV) pH5.5	EE* (%)
Type-D	99	-1.9	33.0	99

*EE; Encapsulation Efficiency

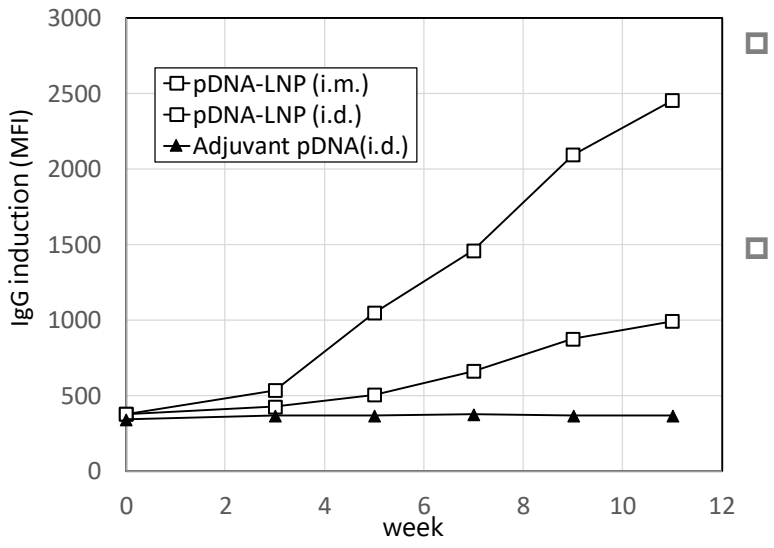
(b)
IgG induction (MFI)
3000
2500
2000
1500
1000
500
0
0
2
4
6
8
10
12
week
pDNA-LNP (i.m.)
pDNA-LNP (i.d.)
Adjuvant pDNA(i.d.)

- Our pDNA-LNPs induced higher IgG titer than adjuvanted-pDNA complex when administered intramuscularly and intradermally.
- The following studies are ongoing:
 -Intravenous administration (hepatocyte)
 -Elucidation of key structural, biochemical and physicochemical properties for DNA transfection.

Figure 3. pDNA delivery and IgG induction in Rabbits

(a) Administration protocol of test substances, i.m. or i.d., 200ug/head, N = 2 (b) IgG induction of pDNA-LNP(i.m. or i.d.) or commercially available immunization reagent complexed with pDNA(i.d.).

Fujifilm CDMO service

For customers at research phase

✓Original lipids including FL-2266※ and FL-0445※ (Patent granted in JP)
- high expression, rapidly metabolized, and applicable to mRNA

✓Formulation development and optimization from discovery to clinical

※We established a sub-kilogram scale synthesis process and completed GMP production.

For customers at development phase

✓Microfluidic mixing system (NanoAssemblr® system)
- Scalable & reproducible (upto GMP manufacturing)
- Supported by Precision Nanosystems through strategic alliance
- Analytical services including the development of test methods

- The contribution of formulation technology throughout development phases

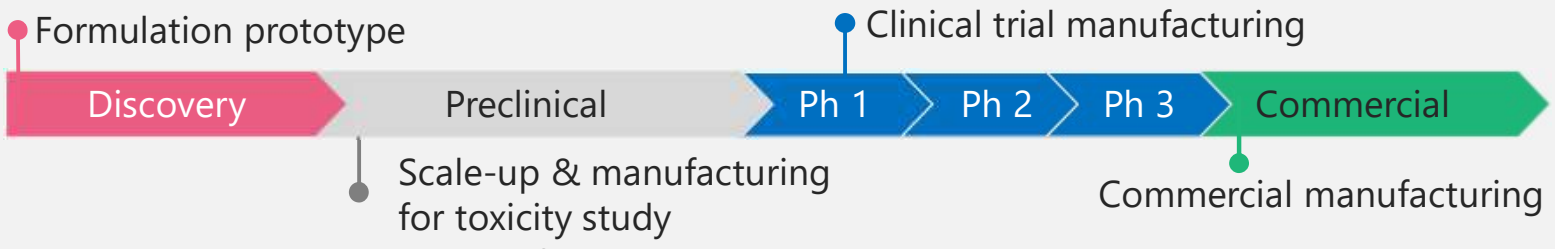
Formulation prototype
Clinical trial manufacturing
Discovery
Preclinical
Ph 1
Ph 2
Ph 3
Commercial
Scale-up & manufacturing
for toxicity study
Commercial manufacturing

- Seamless scale-up manufacture using NanoAssemblr® system

Please Visit Our Booth: **No. 11**

Our Website & LinkedIn：

https://www.fujifilm.com/jp/ja/business/cdmo/medical/liposome

CDMO website

LinkedIn