2615 A Phase 2a Safety Run-in and Preliminary Efficacy Study Of Liposomal Gemcitabine (FF-10832) in Combination with Pembrolizumab in Patients with Advanced Solid Tumors

C. Langer[1], B. Curti[2], C. Farber[3], C. Wheeler[4], D. Wages[4], T. Matsumoto[5], S. Shimoyama[4], M. Mori[5], N. Yamada[4], R.A. Subach[4], T. Madden[4], M. Johansen[4], G. Maier[4], K. Cheung[4] , M. Chaney[6], M. Galsky[7]

Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA[1], Providence Portland Medical Center, Portland, OR[2], Atlantic Hematology Oncology, Morristown Medical Center, Morristown, NJ[3], FUJIFILM Pharmaceuticals U.S.A., Inc., Cambridge, MA[4], FUJIFILM Corporation, Tokyo, Japan[5], Merck & Co., Inc., Rahway, NJ. [6], Icahn School of Medicine at Mount Sinai, New York, NY[7]

This study is in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA

Liposomal Encapsulation Provides Stable, Consistent Delivery of Gemcitabine

FF-10832 - a Novel Liposomal Formulation of Gemcitabine

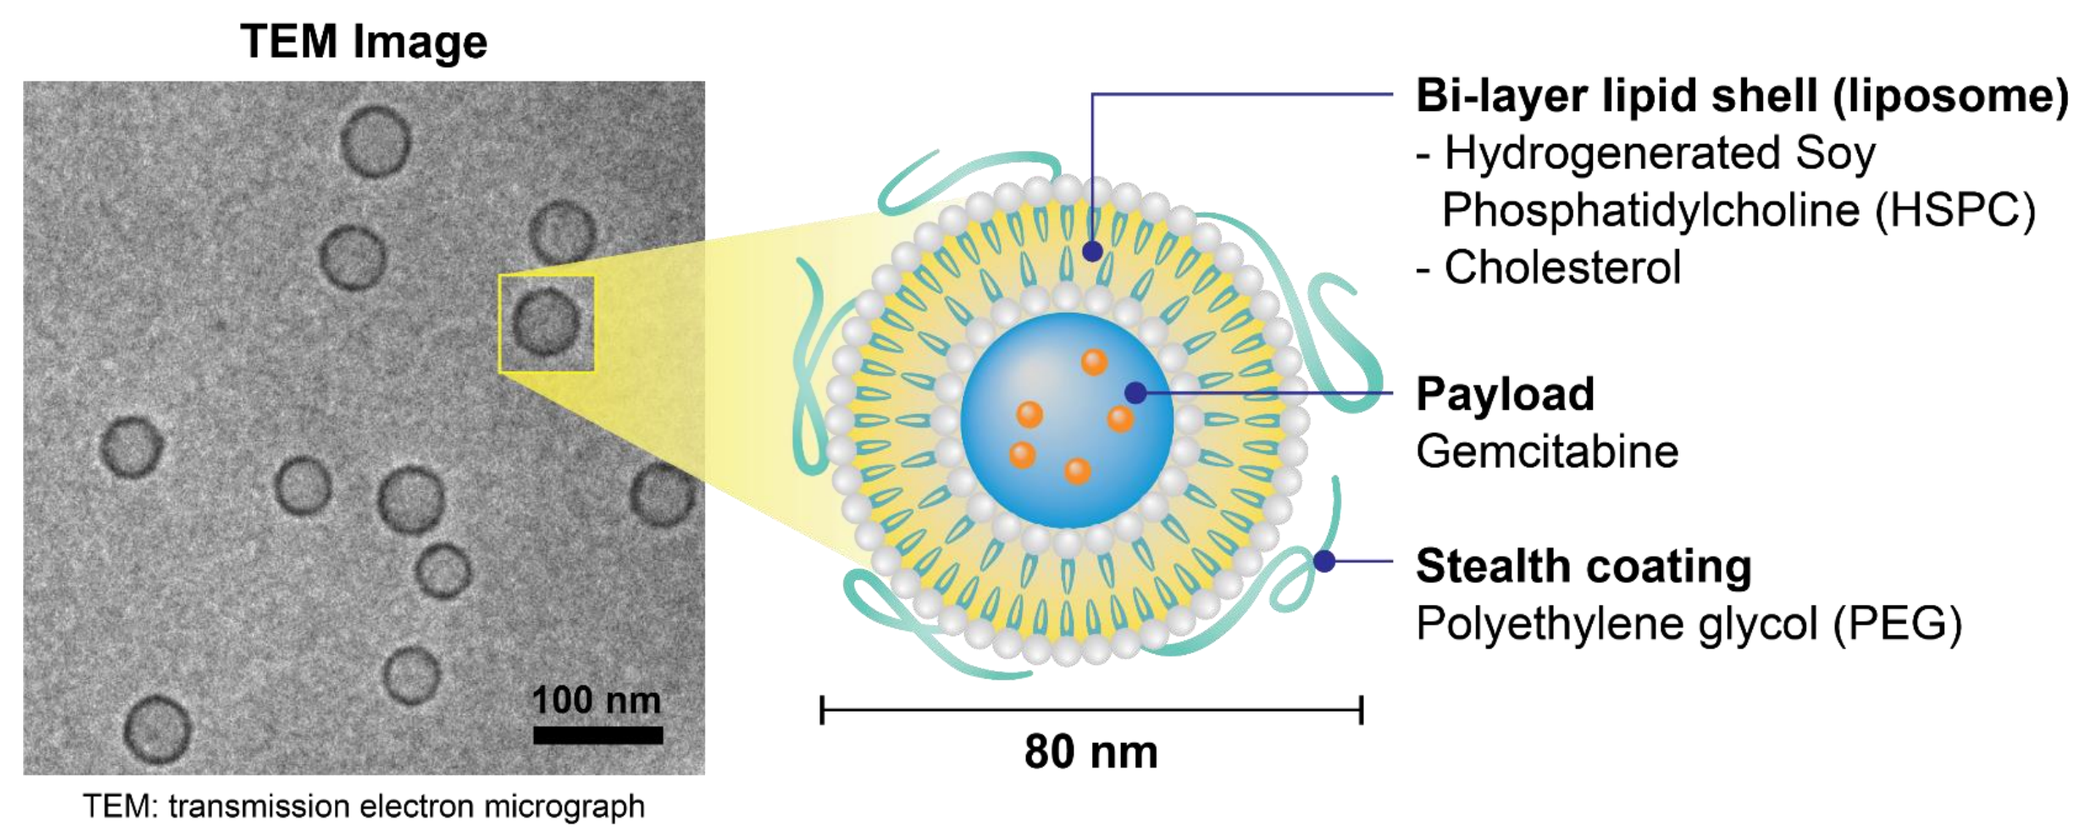
TEM Image
100 nm
80 nm
Bi-layer lipid shell (liposome)
- Hydrogenerated Soy Phosphatidylcholine (HSPC)
- Cholesterol
Payload
Gemcitabine
Stealth coating
Polyethylene glycol (PEG)

TEM: transmission electron micrograph

- A Phase 1 dose escalation study was conducted in patients with advanced solid tumors (NCT03440450)[1]; the recommended phase 2 dose was determined to be 40 mg/m^2 q 21 days

FF-10832 + Anti-PD-1 Has Increased Anti-tumor Activity

Inoculation: EMT6 breast cancer cell
(1x10⁶/mouse, Balb/cCrl Crlj, ♀, 5wk age); N=8 for all groups

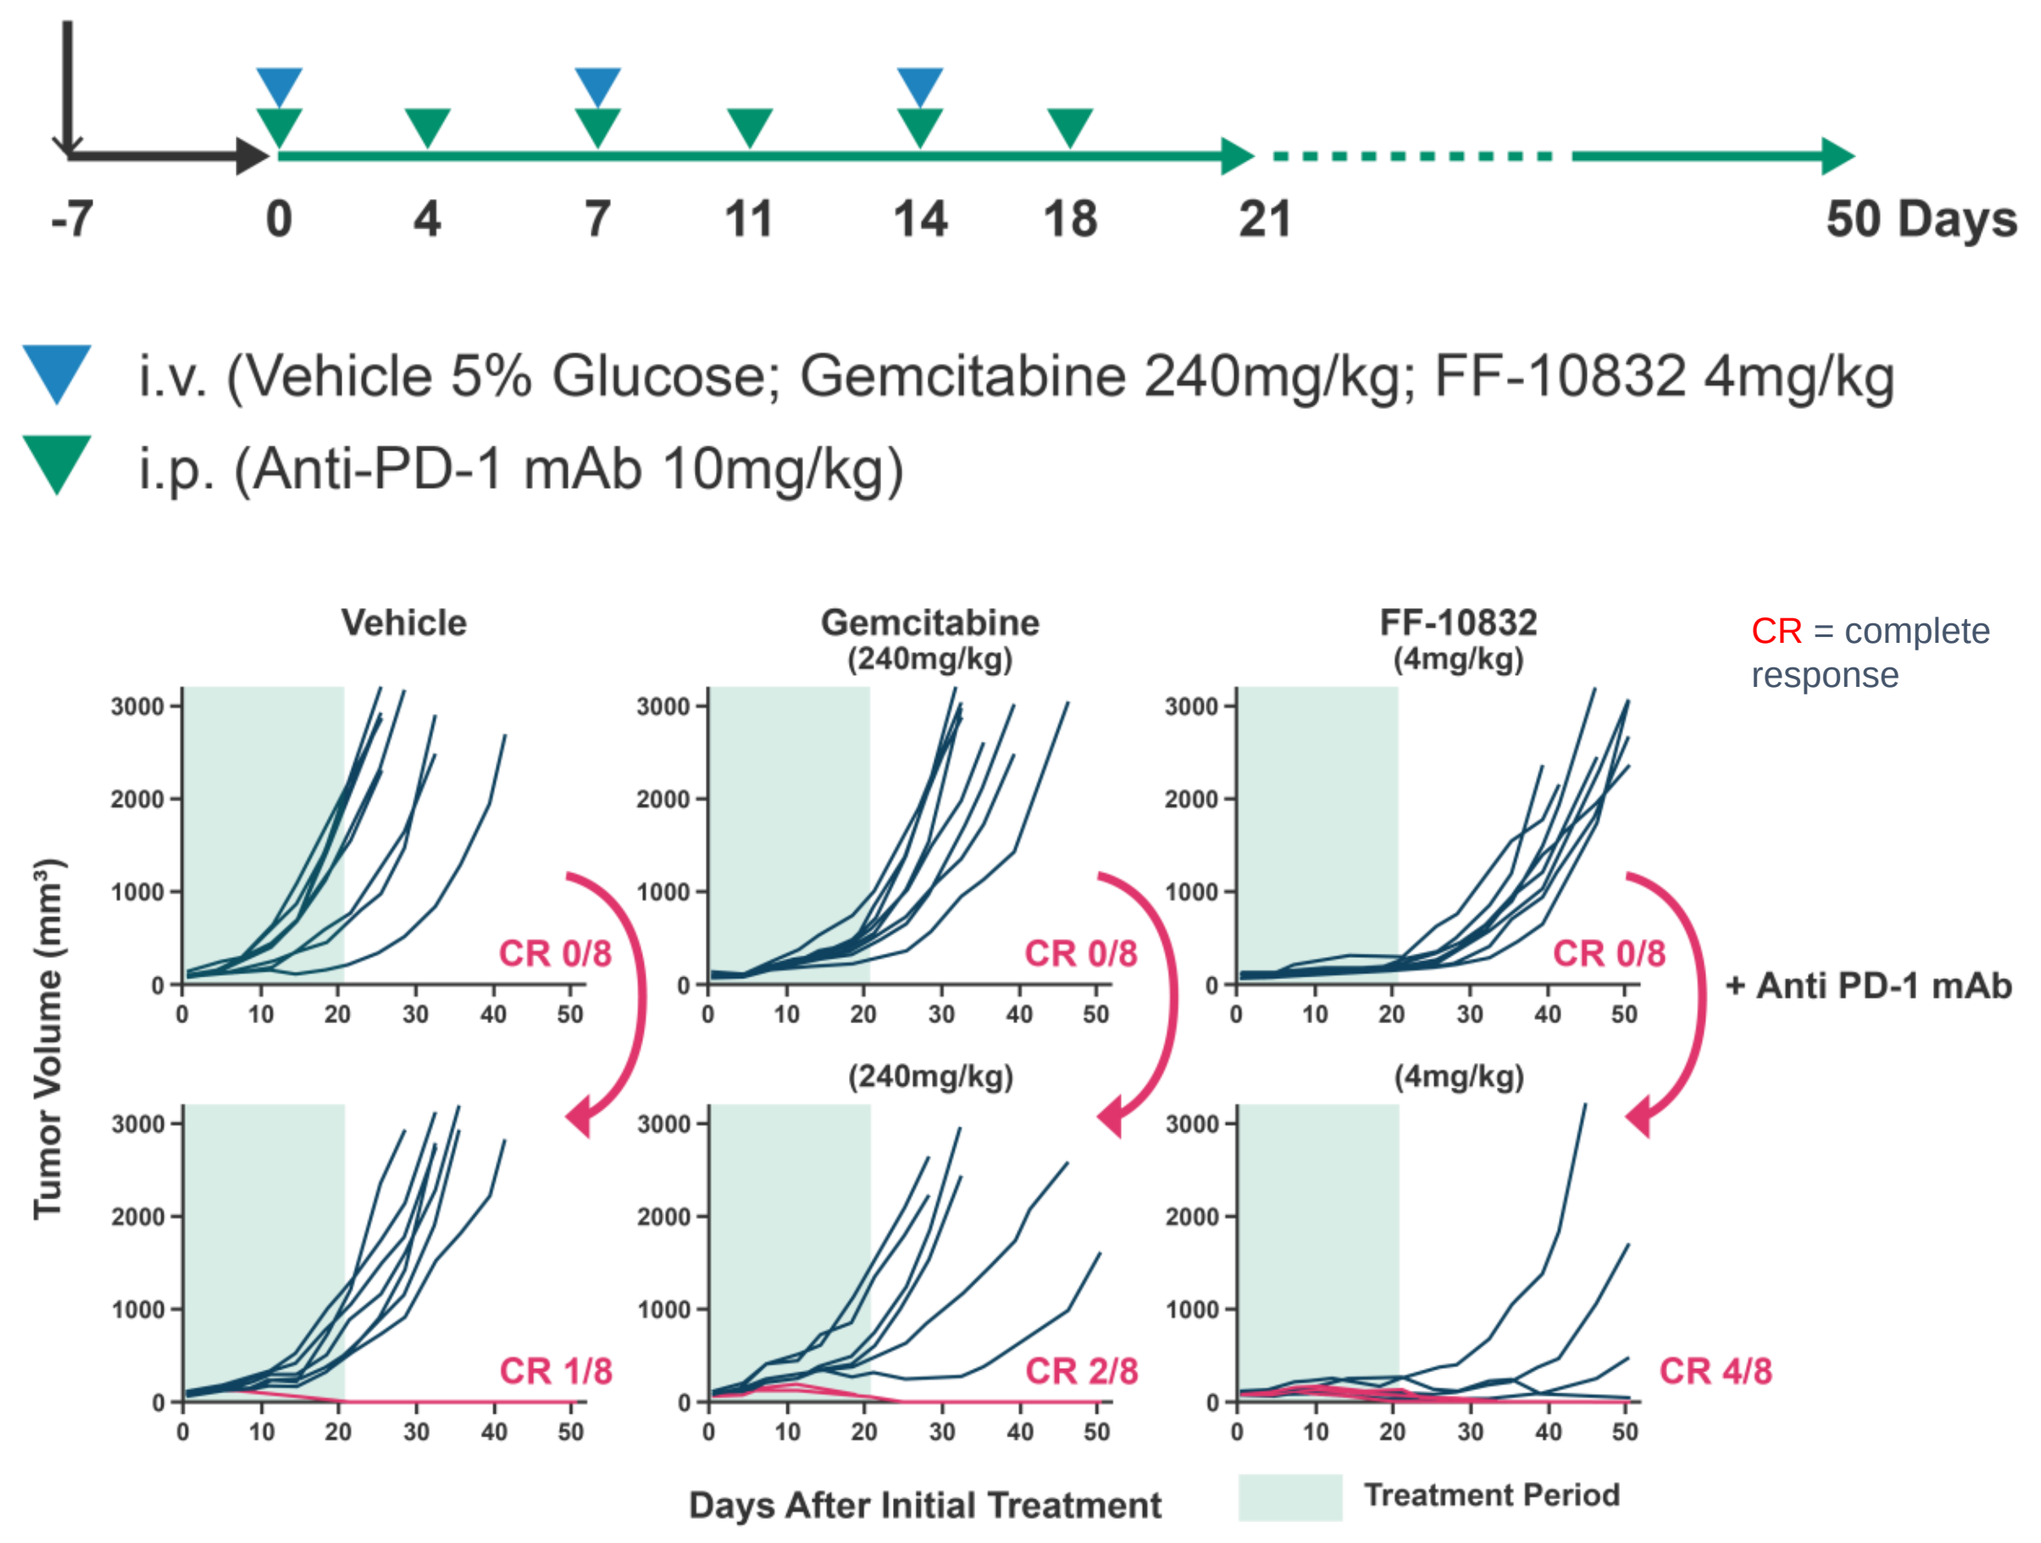
-7
0
4
7
11
14
18
21
50 Days
i.v. (Vehicle 5% Glucose; Gemcitabine 240mg/kg; FF-10832 4mg/kg
i.p. (Anti-PD-1 mAb 10mg/kg)
Vehicle
Gemcitabine (240mg/kg)
FF-10832 (4mg/kg)
CR = complete response
Tumor Volume (mm³)
CR 0/8
CR 0/8
CR 0/8
+ Anti PD-1 mAb
(240mg/kg)
(4mg/kg)
CR 1/8
CR 2/8
CR 4/8
Days After Initial Treatment
Treatment Period

- Preclinical data demonstrate enhanced anti-tumor activity with FF-10832 compared to gemcitabine when combined with an anti-PD-1 monoclonal antibody

Safety Run-in Key Entry Criteria

- ≥ 18 years of age with advanced/metastatic cancer
- No limit on prior lines of therapy
- ECOG status of 0 or 1; Life expectancy of ≥ 3 months
- Hgb ≥9 g/dL; Plts ≥100 K/µL; ANC ≥1.5 K/µL
- Creatinine and bilirubin ≤1.5X ULN; AST/ALT ≤2.5X ULN (≤5X ULN for patients with hepatic metastases)
- Serious cardiac conditions (e.g. NYHA class III or IV) exclusionary

Study Design: Data Are Presented For Safety Run-in Phase (Cohort 1)

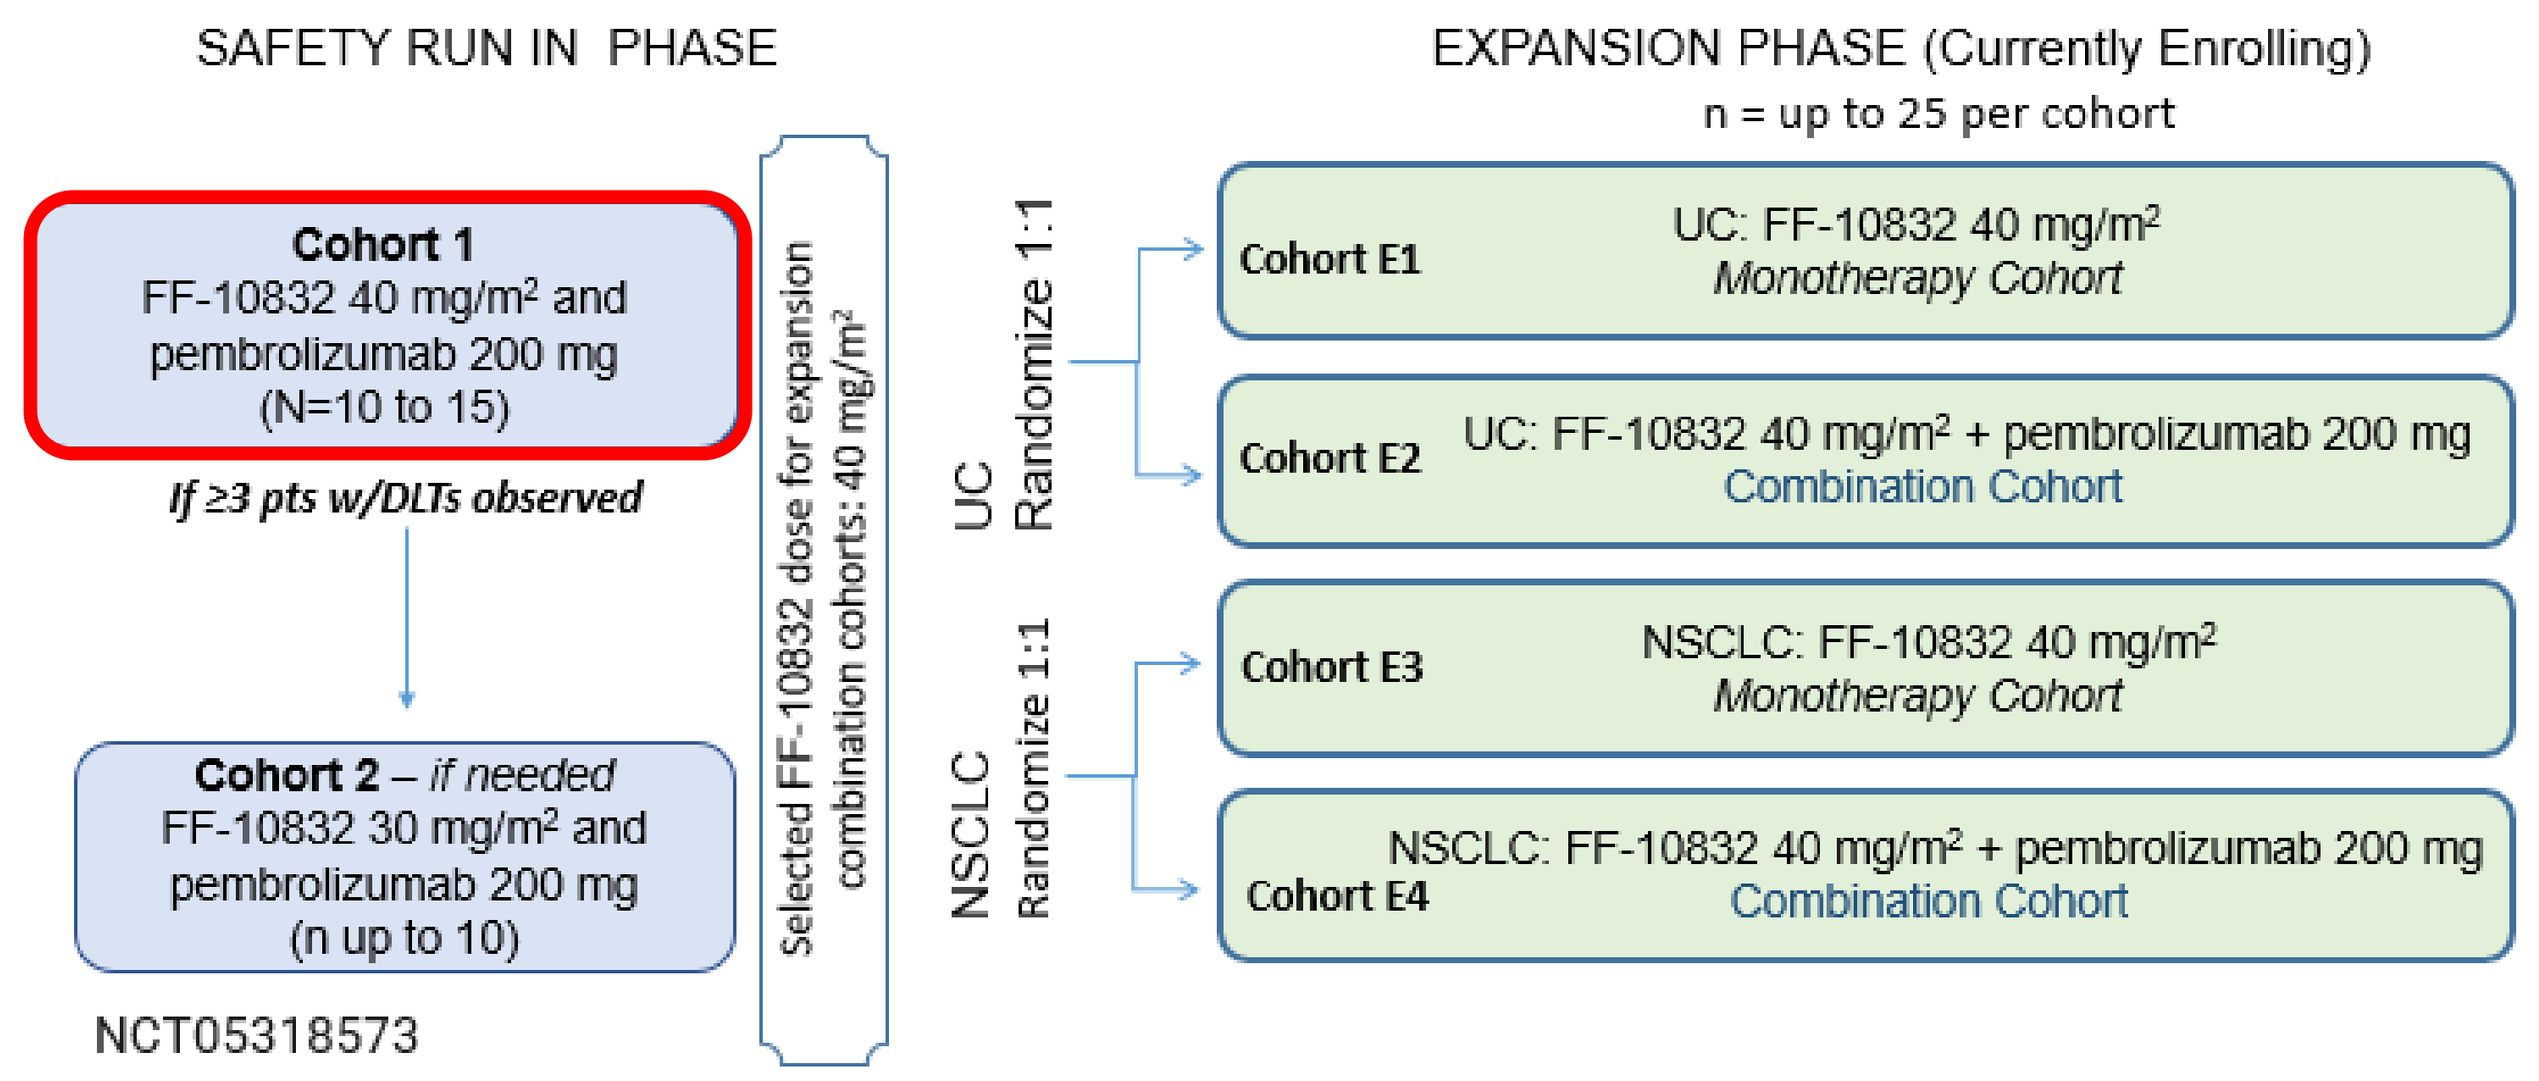
SAFETY RUN IN PHASE
EXPANSION PHASE (Currently Enrolling)
n = up to 25 per cohort
Cohort 1
FF-10832 40 mg/m² and pembrolizumab 200 mg
(N=10 to 15)
If ≥3 pts w/DLTs observed
Cohort 2 – if needed
FF-10832 30 mg/m² and pembrolizumab 200 mg
(n up to 10)
NCT05318573
Selected FF-10832 dose for expansion combination cohorts: 40 mg/m²
UC Randomize 1:1
NSCLC Randomize 1:1
Cohort E1
UC: FF-10832 40 mg/m²
Monotherapy Cohort
Cohort E2
UC: FF-10832 40 mg/m² + pembrolizumab 200 mg
Combination Cohort
Cohort E3
NSCLC: FF-10832 40 mg/m²
Monotherapy Cohort
Cohort E4
NSCLC: FF-10832 40 mg/m² + pembrolizumab 200 mg
Combination Cohort

- FF-10832 & pembrolizumab administered on Day 1 of a 21-day cycle
- Treatment continued until unacceptable toxicity or disease progression
- RECIST 1.1 and iRECIST evaluation performed every 2 cycles
- DLT defined per Phase 1 FF-10832 monotherapy trial[1]

Baseline Demographics

Baseline demographics (n=12 treated patients)	
Median age, years (range)	69 (42–82)
Male/female, n	6 / 6
Primary cancer, n NSCLC Urothelial carcinoma Renal cell carcinoma	 6 4 2
ECOG performance status, n, (0 / 1)	2 / 10
Prior cancer therapy Number of prior regimens, median (range) Best response to most recent therapy, n	 5 (1 - 7) PR(1), SD(4), PD(6), UNK(1)
Prior gemcitabine therapy, n (%) Best response to prior gemcitabine, n	5 (42%) PR(2), SD(1), PD(1), UNK(1)
Prior PD-1/PD-L1 therapy, n (%) Best response to prior PD-1/L1 therapy, n	12 (100%) PR(2), SD(5), PD(4), UNK(1)

Patient Disposition

- All patients have discontinued treatment:
 - 9 patients had disease progression by either radiography (8) or clinical progression (1)
 - 1 patient died during treatment of an unrelated cardiac event*
 - 1 patient discontinued due to physician recommendation**
 - 1 patient discontinued due to grade 2 pyrexia related to FF-10832***

*82 yo M with history of hypertension and stroke. Four days after the first infusion was admitted for hypotension. Hospitalization was complicated by COVID infection with progressive decline ending in cardiac arrest (Patient 5)

**65 yo M who developed a new lesion at the end of Cycle 2 which was considered disease progression requiring a change in therapy (Patient 4)

***69 yo M who developed grade 2 fever, which in context of other unrelated adverse events of weakness and atrial fibrillation prompted removal from study (Patient 10)

Dose Intensity & Safety of the Combination

- 1 DLT occurred (grade 3 pain, malaise and arthralgia)
- Median (range) cycles received: 2 (1-8)
- 2 patients required FF-10832 dose reduction to 30 mg/m^2 after C1
- 1 patient discontinued FF-10832 due to grade 2 pyrexia
- 1 discontinued pembrolizumab due to grade 3 maculopapular rash
 - Entered study with grade 1 rash due to enfortumab vedotin (immediate prior therapy)
- FF-10832 infusion interruptions/dosing delays:
 - 1 patient had C1D1 dose interrupted due to infusion reaction (facial flushing, HTN, dyspnea, tachycardia); resolved with treatment (steroid/H_2 antagonist) & dosing completed
 - 1 patient had treatment delayed due to an embolic stroke prior to Cycle 6, unrelated to FF-10832; recovered but taken off study due to PD.
- Most common AEs considered related to FF-10832 + pembrolizumab were Gr≤2 fatigue (50%) & nausea (25%)

Treatment-related AEs in ≥3 Patients*

Adverse Event	Related to FF-10832 + pembrolizumab	Related to FF-10832 only
	All patients (n=12), n (%)	
Fatigue	6 (50)	1 (8)
Nausea	3 (25)	0
Anemia	2 (17)	3 (25)
Muscular weakness	2 (17)	2 (17)
AST, increased	1 (8)	2 (17)
Decreased appetite	1 (8)	3 (25)
Diarrhea	1 (8)	2 (17)
Pyrexia	1 (8)	2 (17)

*Columns are mutually exclusive

Grade ≥3 Treatment-related AEs*

Adverse Event	Related to FF-10832 + pembrolizumab	Related to FF-10832 only
	All patients/all related grade ≥3 events (n=12), n (%)	
Amylase, increased	1 (8)	0
Arthralgia	1 (8)**	0
Lipase, increased	1 (8)	0
Maculopapular rash	1 (8)	0
Malaise	1 (8)**	0
Pain	1 (8)**	0
Anemia	0	2 (17)
Fatigue	0	1 (8)

*Columns are mutually exclusive. All events were grade 3 except for one grade 4 lipase elevation that resolved (grade 3 amylase elevation in same patient)

**Dose-limiting toxicity of grade 3 arthralgia, malaise, pain in same patient

Anti-tumor Activity: iRECIST Response

Best Response by iRECIST	n (%)
iCR	0
iPR	1 (10)
iSD	5 (50)
iCPD	3 (30)
iUPD	1 (10)
Not Available	2*

*2 of 12 treated patients were not evaluable for disease response. Patient 10 discontinued treatment due to related grade 2 fever and other unrelated AEs before scheduled scans were performed. Patient 5 died due to a cardiac event prior to scheduled scans.

- A partial response (PR/iPR) was observed in a 69 yo F with urothelial carcinoma (Patient 9)
 - Prior therapy (best response): gemcitabine/cisplatin (SD), pembrolizumab (unknown), and enfortumab (SD)
 - Two target lesions in anterior and posterior right lung; 8 non-target lesions in lung (2), bone (3), liver (2) and LN (1)
- Median PFS was 6 weeks (95%CI: 3.1–NR); median OS was 23.3 weeks (95% CI: 4–- NR)

Radiographic Response / Treatment Duration

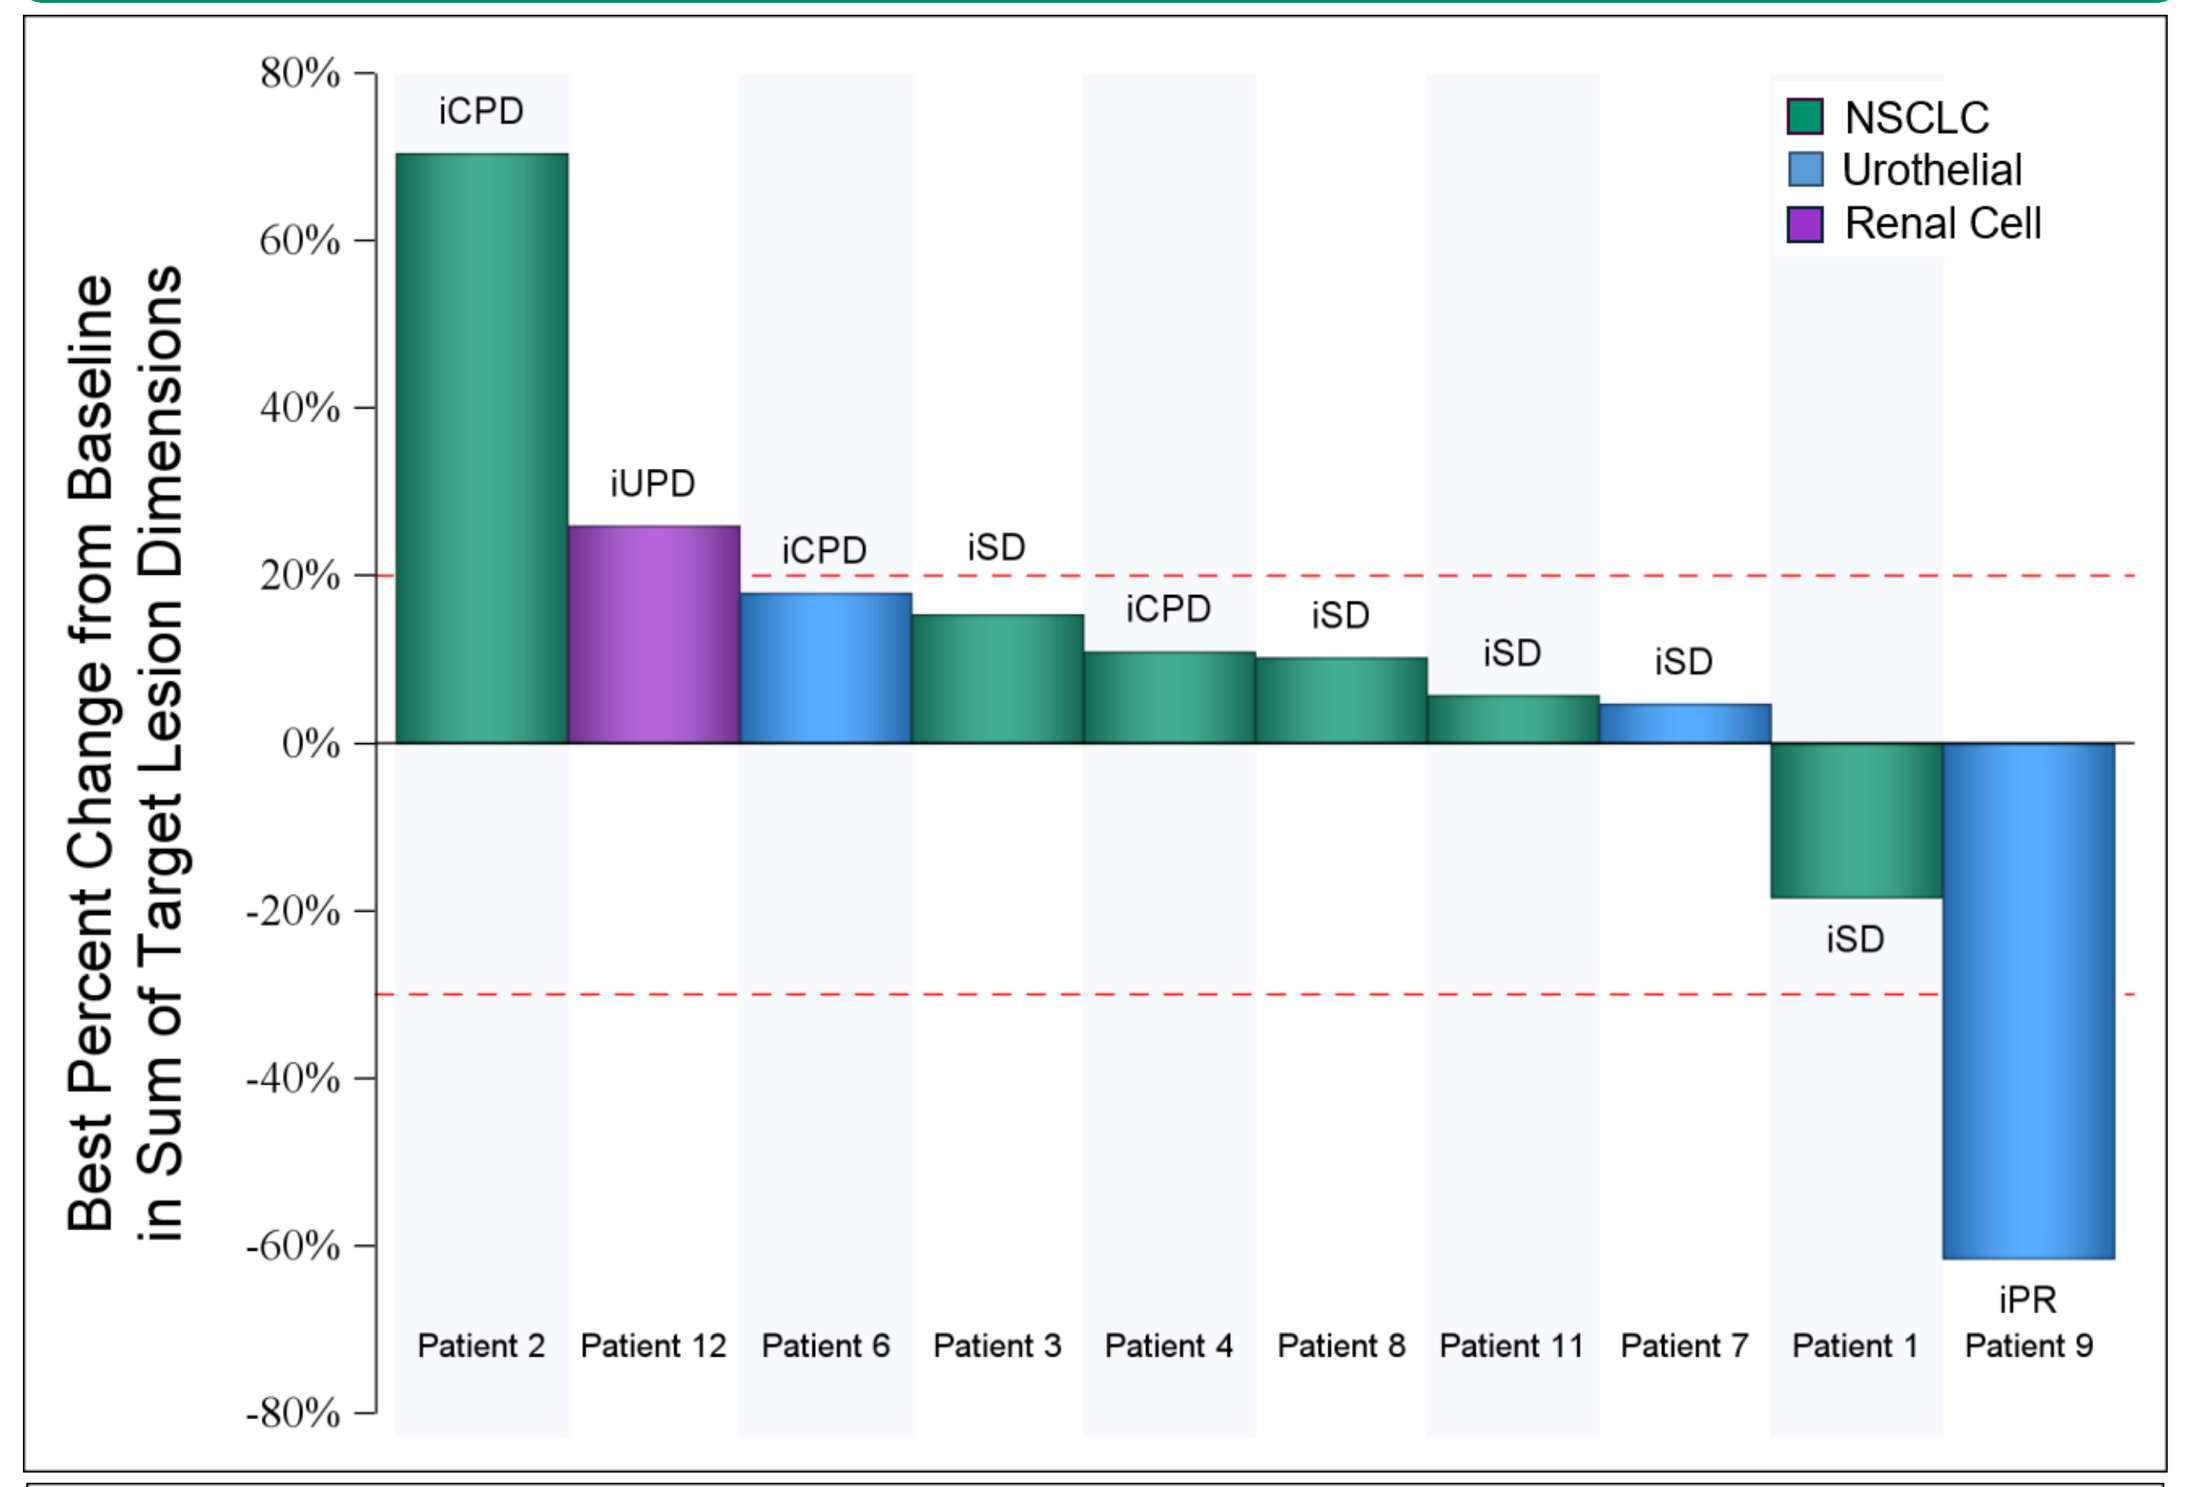
Best Percent Change from Baseline in Sum of Target Lesion Dimensions
80%
60%
40%
20%
0%
-20%
-40%
-60%
-80%
iCPD
iUPD
iCPD
iSD
iCPD
iSD
iSD
iSD
iSD
iPR
Patient 2
Patient 12
Patient 6
Patient 3
Patient 4
Patient 8
Patient 11
Patient 7
Patient 1
Patient 9
NSCLC
Urothelial
Renal Cell

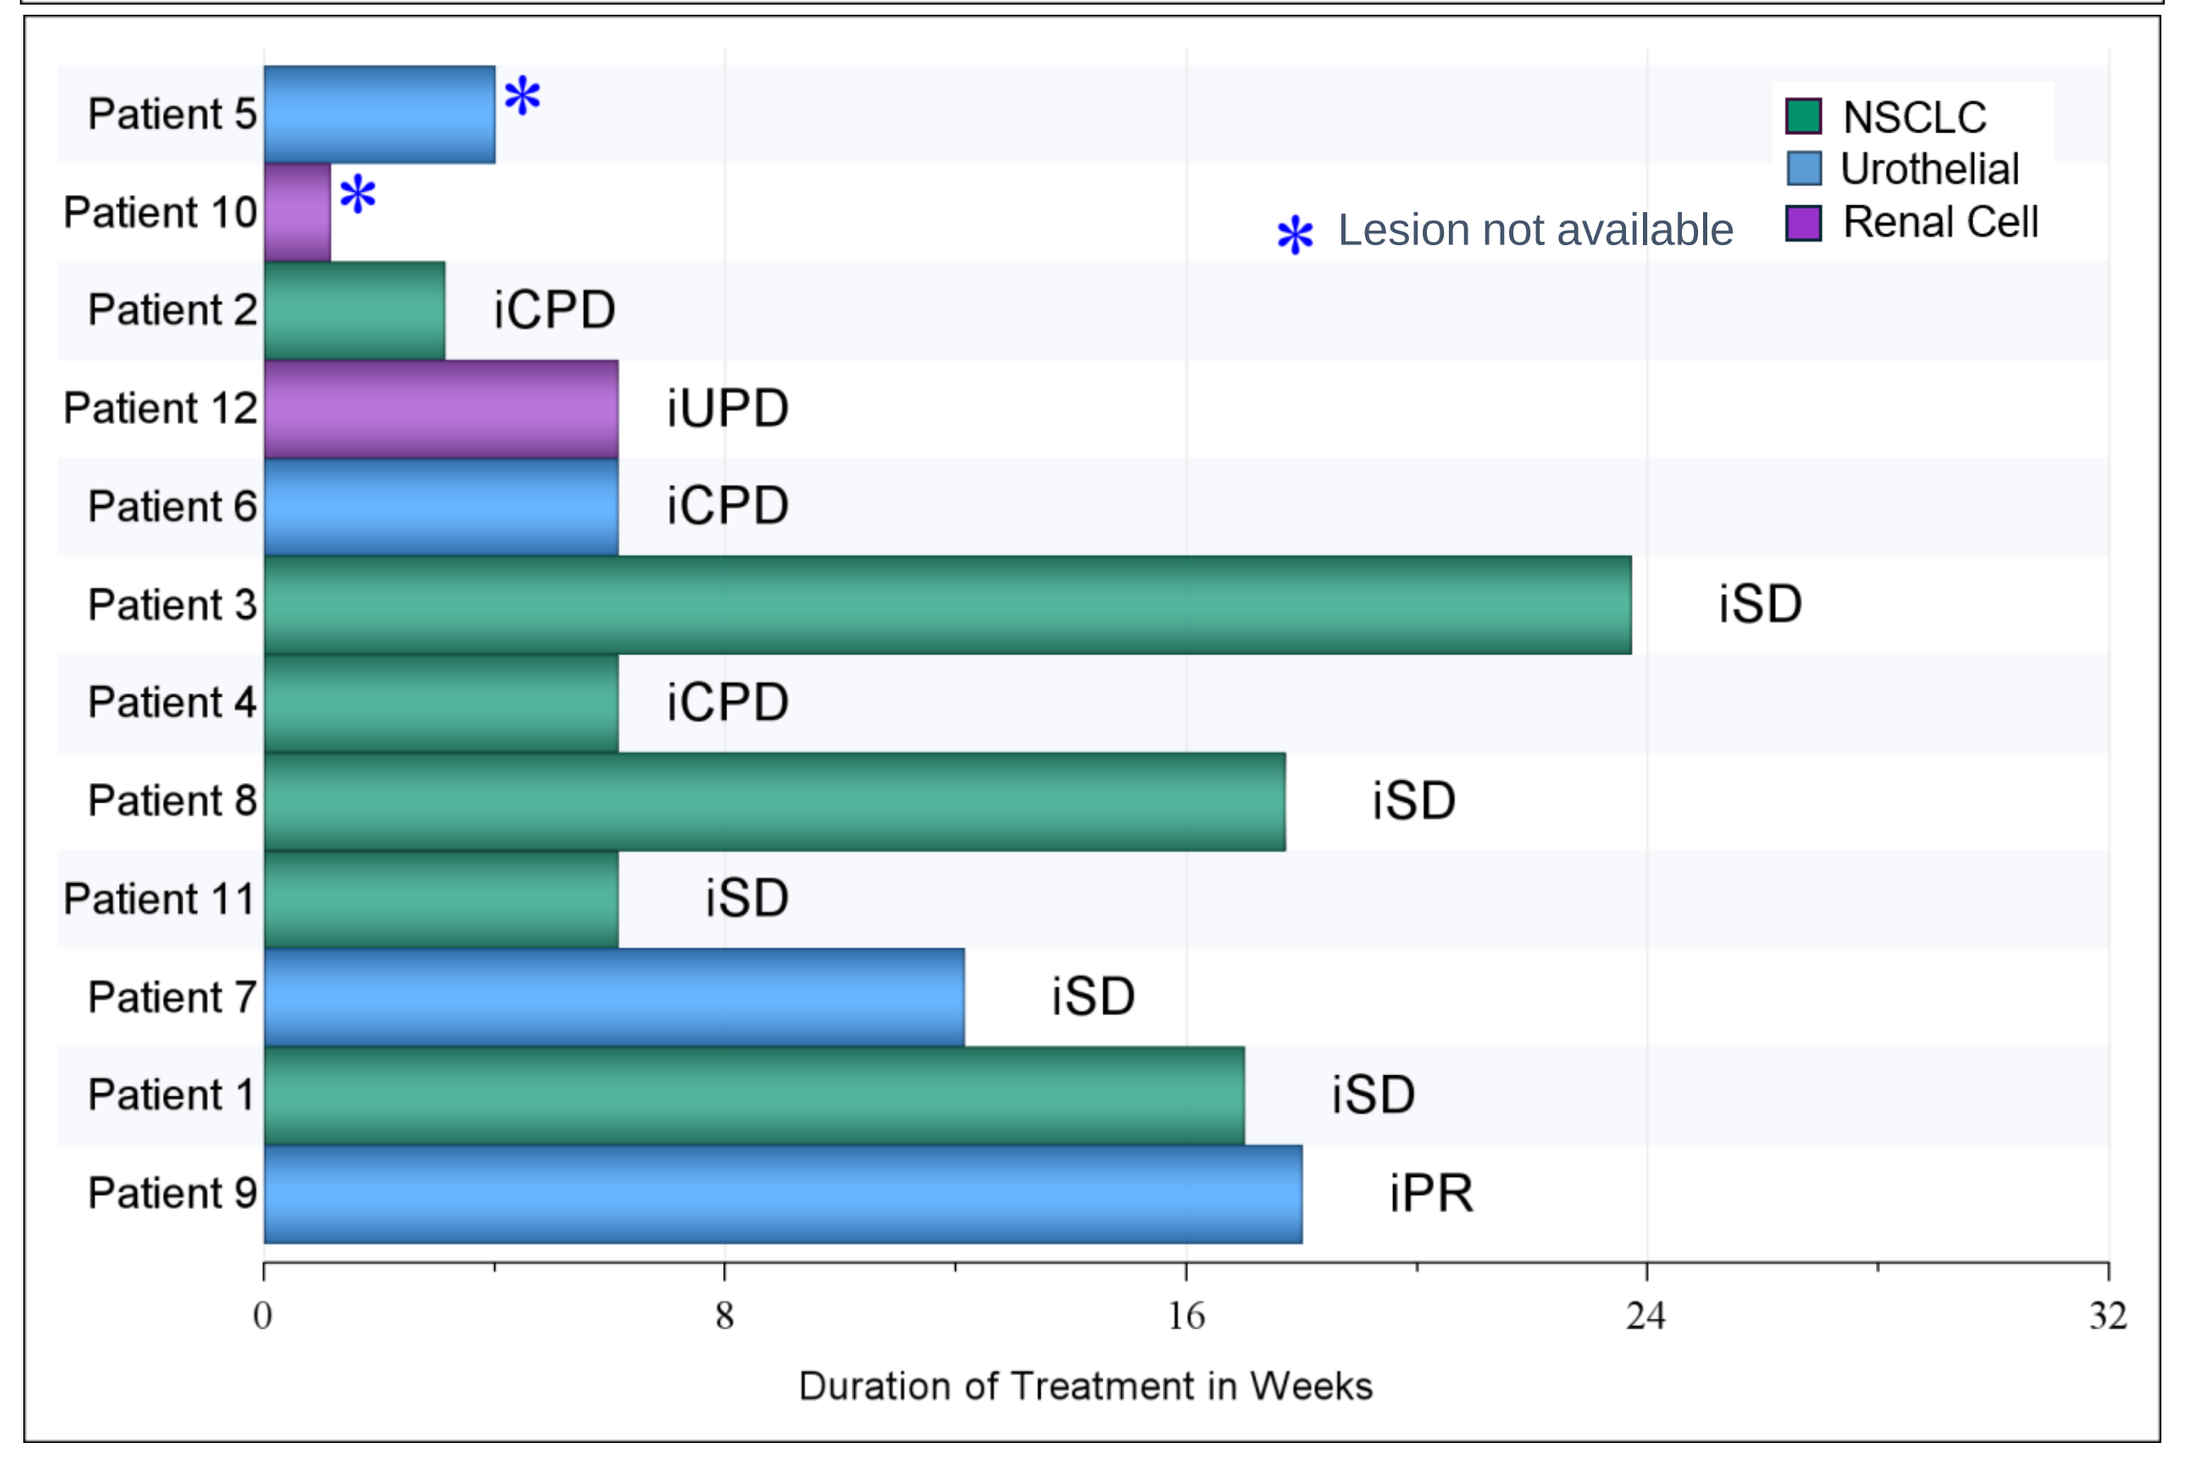
Patient 5
Patient 10
Patient 2
Patient 12
Patient 6
Patient 3
Patient 4
Patient 8
Patient 11
Patient 7
Patient 1
Patient 9
iCPD
iUPD
iCPD
iSD
iCPD
iSD
iSD
iSD
iSD
iPR
Lesion not available
NSCLC
Urothelial
Renal Cell
0
8
16
24
32
Duration of Treatment in Weeks

Peripheral Blood T-Cells

- Circulating T-cells measured as a surrogate of immunocompetency in the tumor microenvironment[2-4]
- While overall population density of Total CD4+ T-cells did not change, 2 - 6-fold log ↓'s were observed in immune suppressive CD4+ Treg lineages in patient achieving PR & some maintaining SD; consistent with Phase 1 FF-10832 monotherapy trial[1]

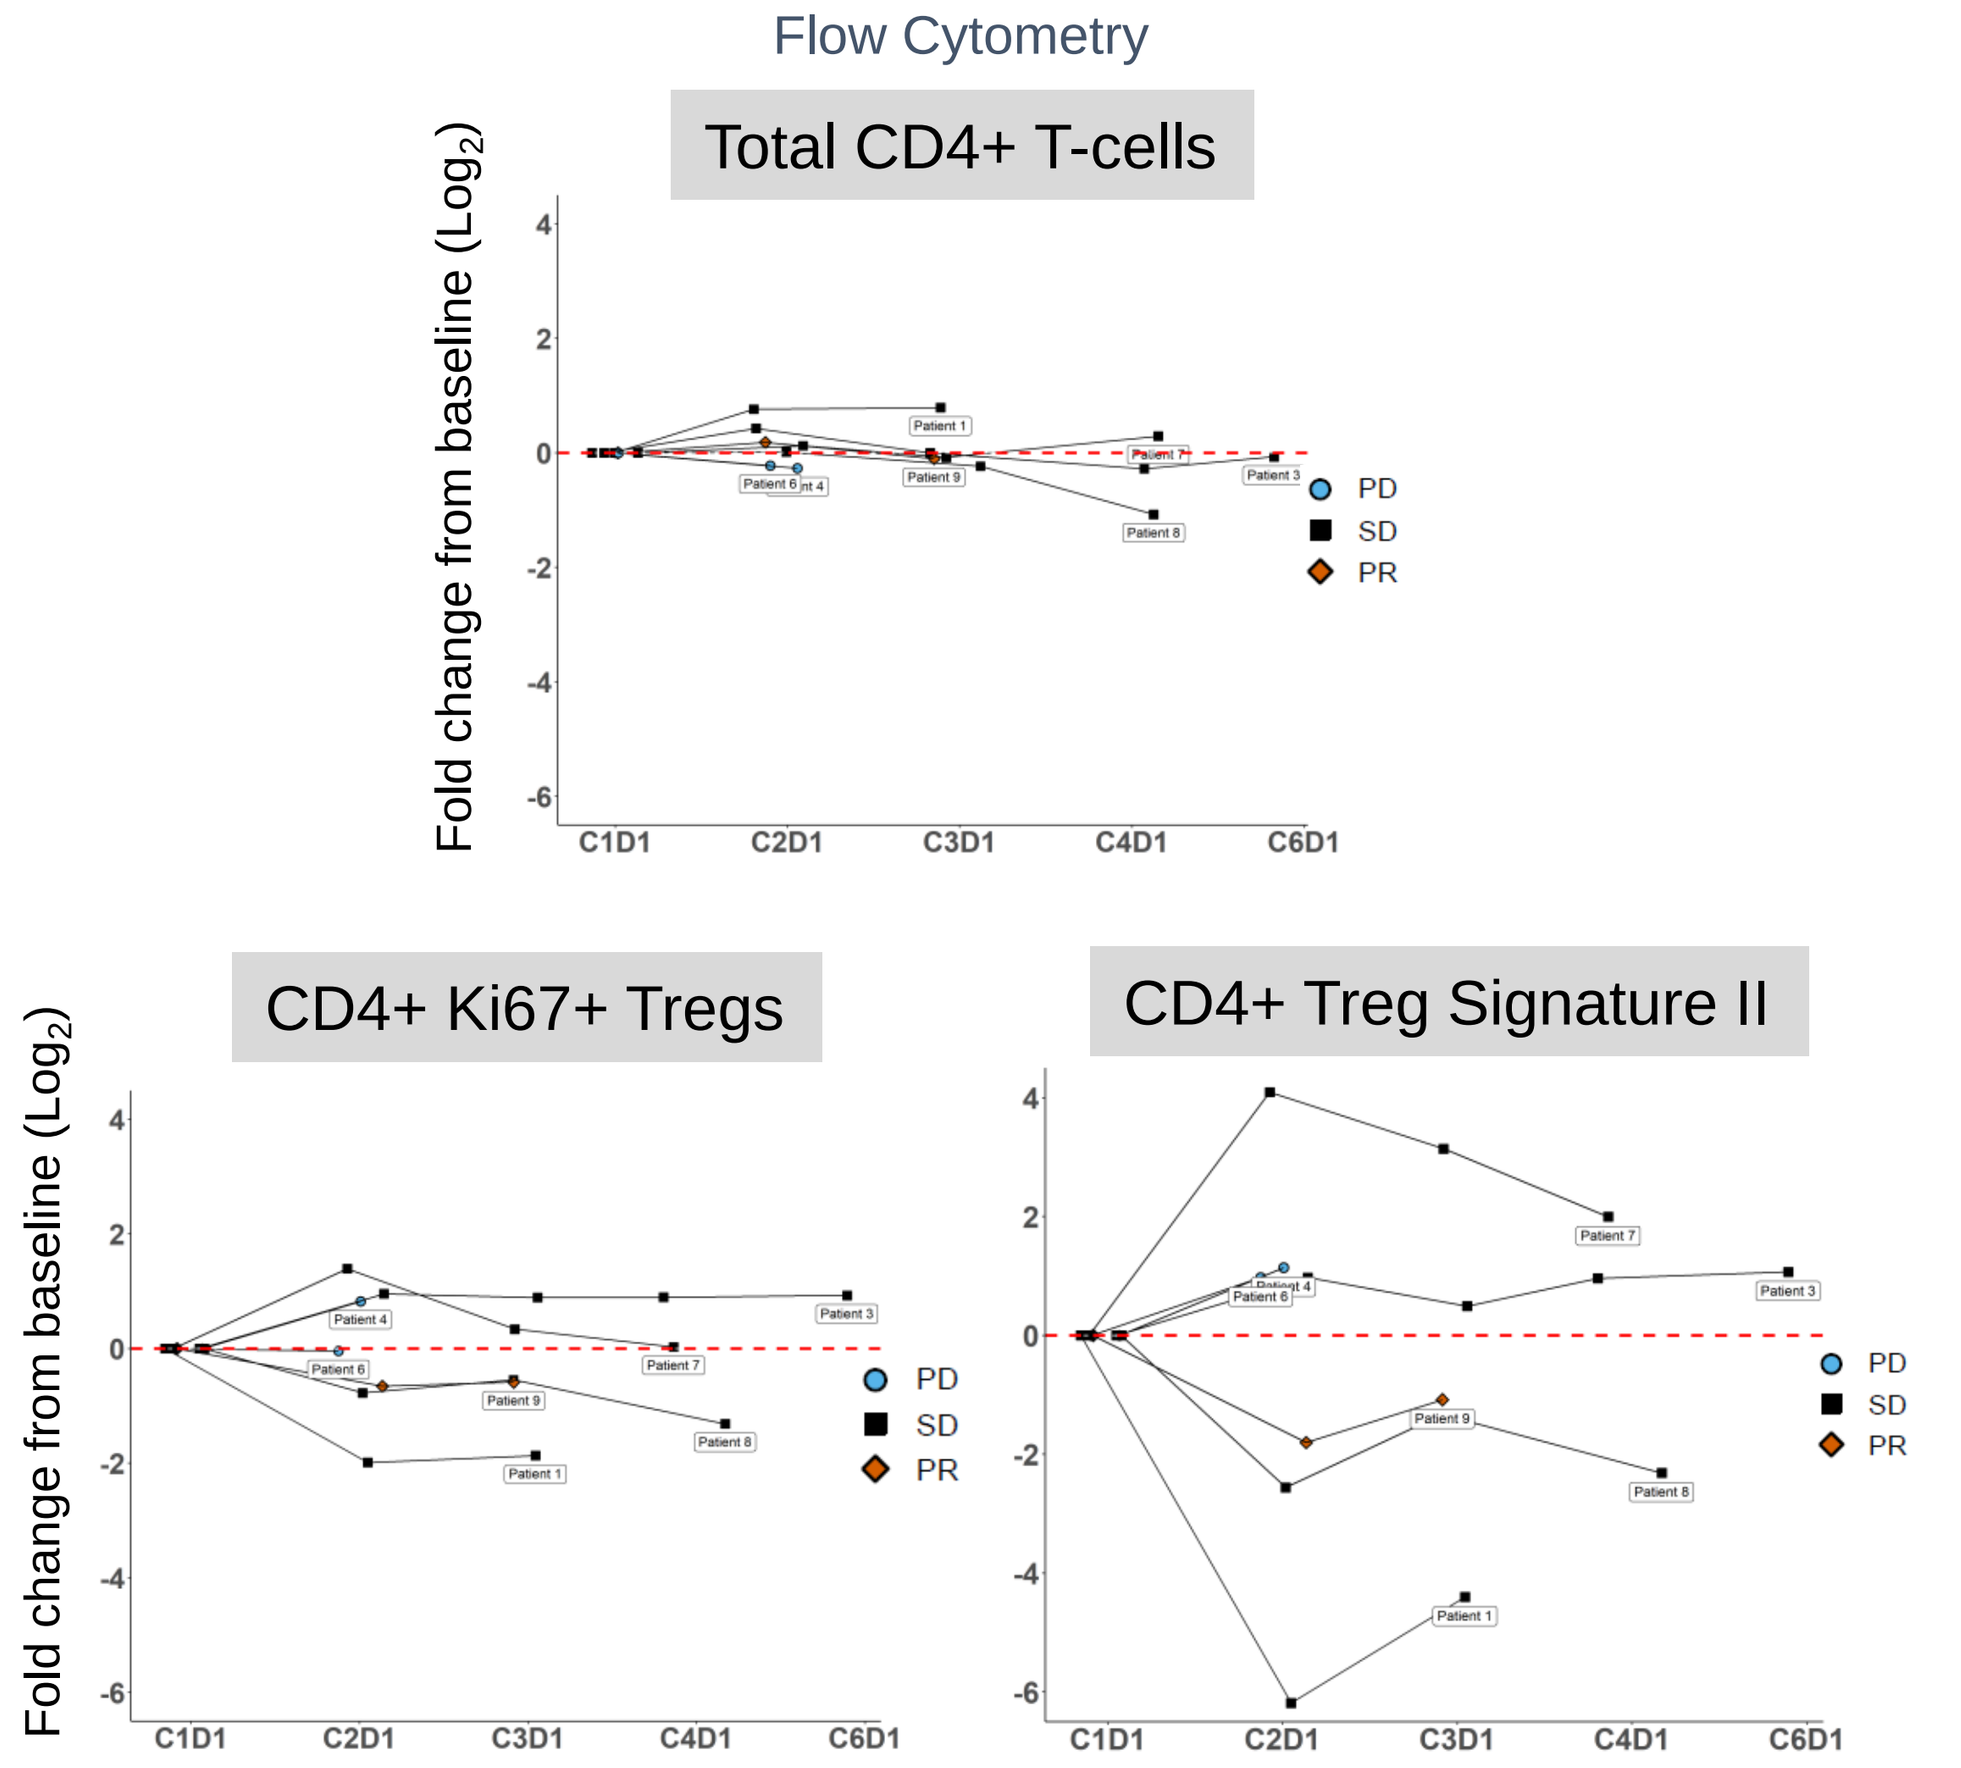
Flow Cytometry
Total CD4+ T-cells
Fold change from baseline (Log_2)
C1D1
C2D1
C3D1
C4D1
C6D1
PD
SD
PR
CD4+ Ki67+ Tregs
CD4+ Treg Signature II

Pharmacokinetics

- PK profile & extended total/free gemcitabine plasma $t_{1/2}$ (~30 hrs) consistent with FF-10832 monotherapy trial[1]

Summary

- The combination of FF-10832 40 mg/m^2 and pembrolizumab 200 mg q 21 days was well tolerated; AEs were consistent with safety profiles for both treatments
- Preliminary evidence of anti-tumor activity was demonstrated in a heavily pre-treated population
- PD analyses suggest potential immunomodulatory effects
- Expansion cohort enrollment ongoing in urothelial and 2L non-small cell lung carcinoma

References

1. *J Clin Oncol* 2022; 40(16-suppl):3097
2. *Nature, Sci Reports* 2021; 11:14426
3. *J Immuno Cancer* 2022; 10(6):1
4. *Cancer Med* 2023; 12(8):9069

Acknowledgements

Sponsor: FUJIFILM Pharmaceuticals U.S.A., Inc.; Support from Translational Drug Development (Scottsdale, AZ), study execution; LabCorp (Madison, WI), bioanalysis; CellCarta (Montreal, Canada), immune cell flow cytometry.

Study contact: Corey.Langer@pennmedicine.upenn.edu

Partnering activities: fphucontact@fujifilm.com

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from ASCO or the author of poster.